Retrospective analysis of appropriate biomarker screening testing prior to treatment for members with HR+/HER2- metastatic breast cancer and real-world treatment pattern

Harvard Huynh, PharmD Candidate 2022, St. John's University, New York – AMCP Foundation/Pfizer, Inc. Summer Oncology Intern, Envolve Pharmacy Solutions (Primary Presenter)
Rodney Gedey, PharmD – Envolve Pharmacy Solutions – Director of Drug Information Services (Co-Author)

AMCP Foundation

envolve™ Pharmacy Solutions

INTRODUCTION

A 2021 American Society of Clinical Oncology annual meeting presentation showed the underuse of biomarker testing in non-small cell lung cancer.

This retrospective cohort study examines whether this is occurring in hormone receptor-positive/human epidermal growth factor receptor 2-negative (HR+/HER2-) metastatic breast cancer (mBC).

HR+/HER2- breast cancer is the most common subtype of breast cancer that accounts for roughly 68% of all subtypes.[1,2]

Immunohistochemistry (IHC) is a biomarker test used to identify HR+/HER2- mBC and the National Comprehensive Cancer Network (NCCN) guidelines recommend HR testing by IHC performed on any newly metastatic breast cancer patient.[2]

The NCCN guidelines recommends CDK4/6 inhibitors (CDK4/6i), Aromatase inhibitors (AI), Antiestrogen, and Estrogen Receptor Down-Regulators (ERD) in combination or individually as a preferred treatment for HR+/HER2- mBC.[2]

Evidence for an appropriate screening test before receiving treatment for HR+/HER2- mBC is limited.

OBJECTIVES

The primary objective is to determine if members with HR+/HER2- mBC are screened appropriately before receiving a CDK4/6i, AIs, Antiestrogen, or ERD.

The secondary outcomes will compare the total medical and total pharmacy costs between the screened and unscreened groups.

METHODS

Study Design

- Retrospective and observational cohort study of HR+/HER2- mBC patients who initiated the selected HR+/HER2- mBC treatments between 6/1/2019 – 6/1/2021 using pharmacy, medical, and laboratory claims.
- Look-back Period: 6 months from initial claim of HR+/HER2- mBC treatment.
- At least one (1) medical or pharmacy claim for one of the following used during the study period:
 - CDK4/6i: Palbociclib (Ibrance®), Ribociclib (Kisqali®), Abemaciclib (Verzenio®), Ribociclib/letrozole (Kisqali Femara®)*
 - AI: Anastrazole (Arimidex®)†, Letrozole (Femara®)†, Exemestane (Aromasin®)
 - Antiestrogen: Tamoxifen
 - ERD: Fulvestrant (Faslodex®)
- ≥3 months of pharmacy/medical claims data‡

* Ribociclib/letrozole (Kisqali Femara®) excluded from CDK4/6i + ERD group
† Nonsteroidal Aromatase Inhibitors (NSAI)
‡ Inclusion criteria for the secondary outcome

Exclusion Criteria

- At least (1) medical or pharmacy claim during the study period for a drug used to treat HER2+ breast cancer.
- At least (1) medical or pharmacy claim for a CDK4/6i, AI, Antiestrogen, ERD used in the look-back period.
- At least (1) medical or pharmacy claim with any secondary metastatic ICD-10 code past the look back period.

Data Sources

- Centene Corp. medical and pharmacy claims databases.
- Centene is the largest Medicaid managed care organization in the United States.

Statistical Analysis

- Data was analyzed using SAS analytics.
- Chi square test is conducted to determine the statistical significance between the two groups for the primary outcome.
- Student t-test of unequal variances is conducted to determine statistical significance for the secondary outcomes.

LIMITATIONS

- All the pharmacy claims data within the study period was captured, which led to the misc. column being formed.
- All the medical spend data was captured within the study period with no specific lookback period for spend data.
- The study does not include chemotherapy and chemotherapy can account for a large portion of medical spend.
- Claims data may have coding errors and missing data
- Medical claims are not adjudicated in real-time, and some medical claims may not have been reported.
- It is difficult to discern whether providers are following correct guidelines with claims data.

RESULTS

Table 1. Baseline Characteristics

Age Range, n(%)	
18-49	526 (20.4%)
50-59	714 (27.7%)
60-64	565 (21.9%)
65-69	314 (12.2%)
70-74	172 (6.7%)
75-79	118 (4.6%)
>80	168 (6.5%)
Gender, n(%)	
Female	2549 (98.9%)
Male	28 (1.1%)
Line of Business, n(%)	
COMMERCIAL	52 (2.0%)
DUALS	6 (0.2%)
MARKETPLACE	724 (28.1%)
MEDICAID	1091 (42.3%)
MEDICARE	704 (27.3%)

HR+/HER2- mBC population identification (Table 1)

2,577 members met the inclusion criteria for the study and have HR+/HER2- mBC with their initial claim for a CDK4/6i, AI, Antiestrogen, or ERD between June 1, 2019, through June 1, 2021.

The majority of members were between the ages 50-59, female, and are covered under Medicaid.

Figure 1. Percentage of Patients Tested by Treatment Subgroups

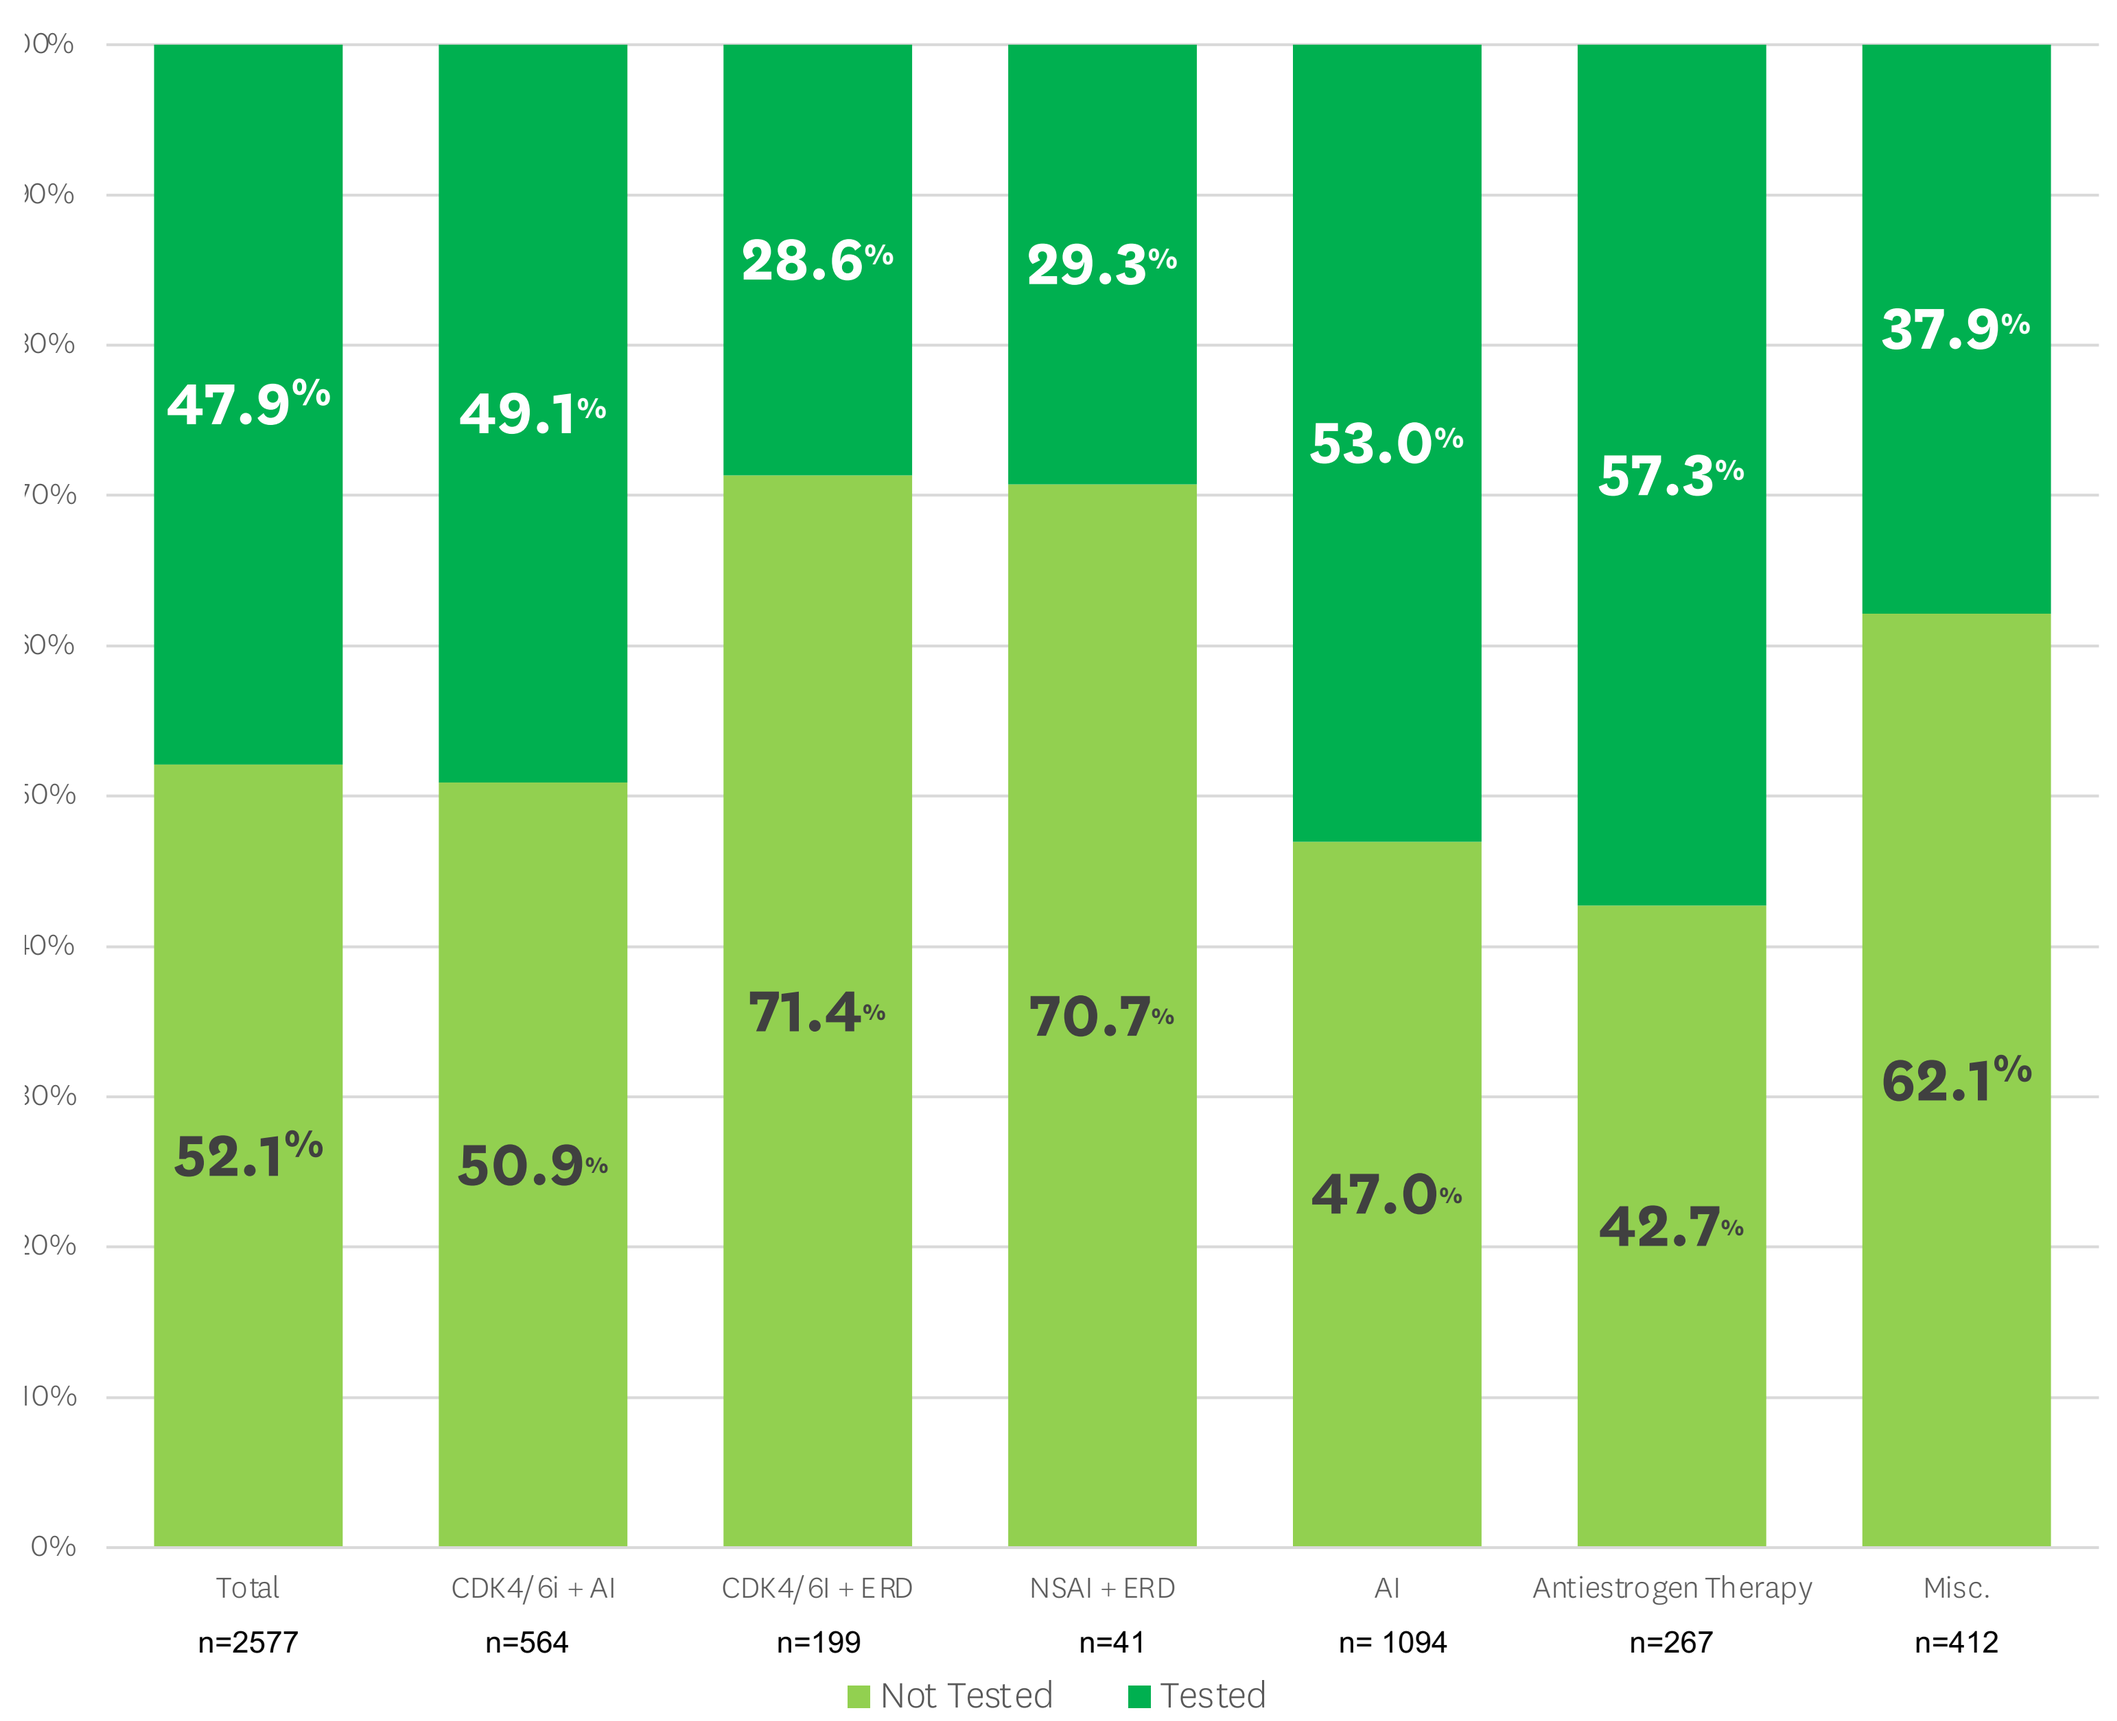
0%
10%
20%
30%
40%
50%
60%
70%
80%
90%
100%
47.9%
52.1%
49.1%
50.9%
28.6%
71.4%
29.3%
70.7%
53.0%
47.0%
57.3%
42.7%
37.9%
62.1%
Total n=2577
CDK4/6i + AI n=564
CDK4/6i + ERD n=199
NSAI + ERD n=41
AI n= 1094
Antiestrogen Therapy n=267
Misc. n=412
Not Tested
Tested

Primary Outcome - Percentage of Patients Tested by Treatment Subgroups (Figure 1)

About 48% of the 2,577 total members received an IHC test prior to their initial claim for a CDK4/6i, AI, Antiestrogen, or ERD (p<.0001)

CDK4/6i + ERD and NSAI + ERD had the largest percentage of untested members with both being roughly 71%.

Figure 2. HR+/HER2- mBC Preferred Treatment Subgroups

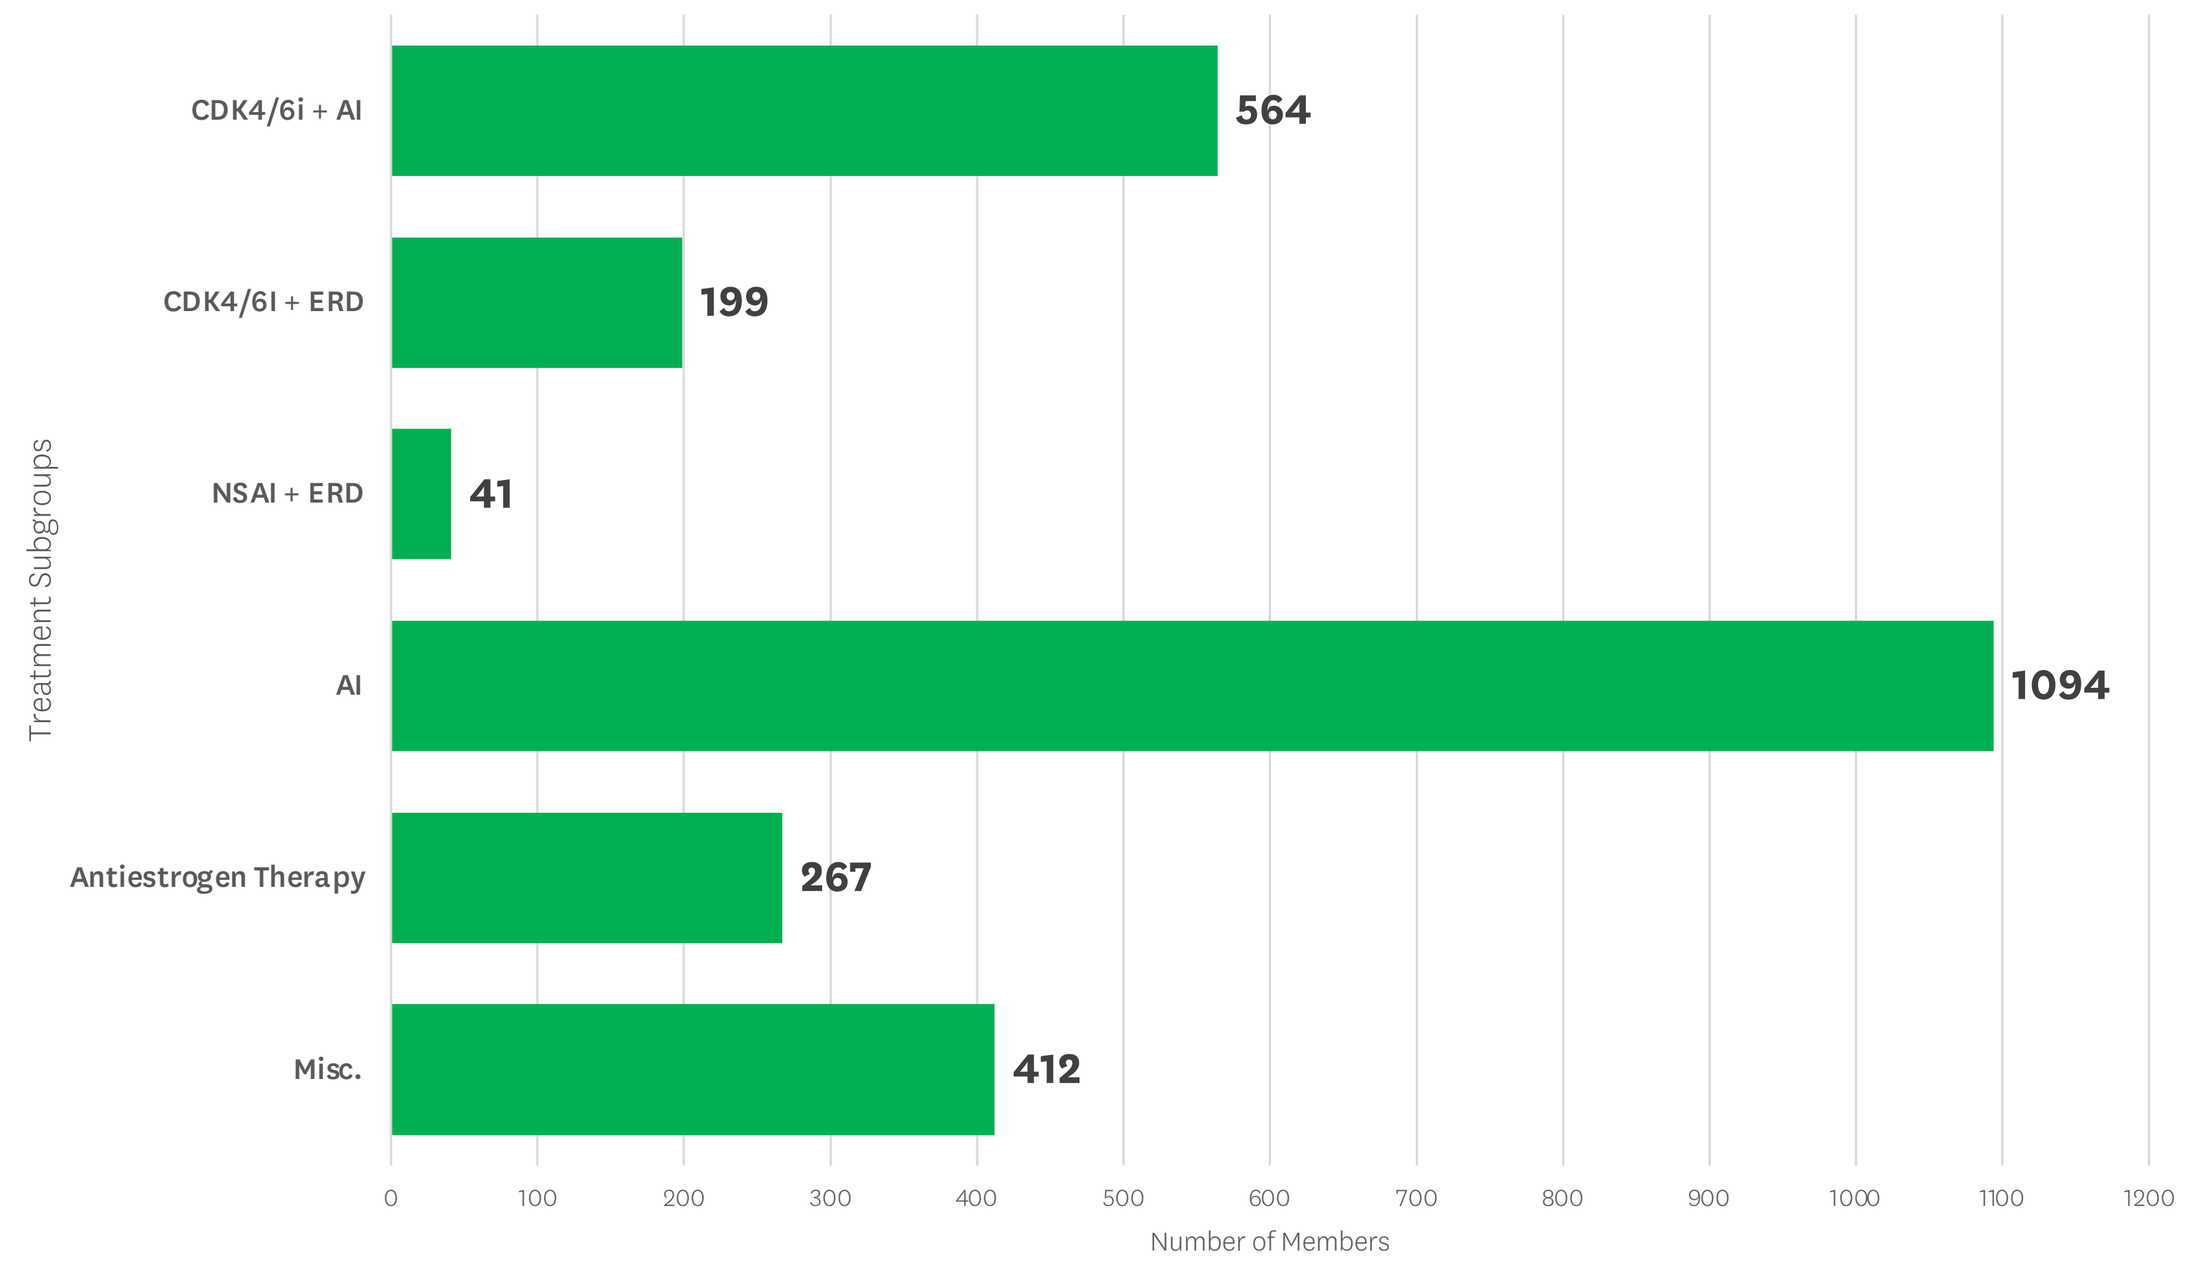
Treatment Subgroups
CDK4/6i + AI
564
CDK4/6i + ERD
199
NSAI + ERD
41
AI
1094
Antiestrogen Therapy
267
Misc.
412
0
100
200
300
400
500
600
700
800
900
1000
1100
1200
Number of Members

HR+/HER2- mBC Preferred Treatment Subgroups (Figure 2)

Majority of members had claims for antiestrogen therapy alone (42%).

Misc. contained treatment combinations that did not fit the following categories, nor were part of the NCCN guidelines. Since all pharmacy claims were captured within the study period and due to the nature of claim data, it is difficult to discern whether providers are following correct guidelines.

Figure 3a. Average Total Medical Spend

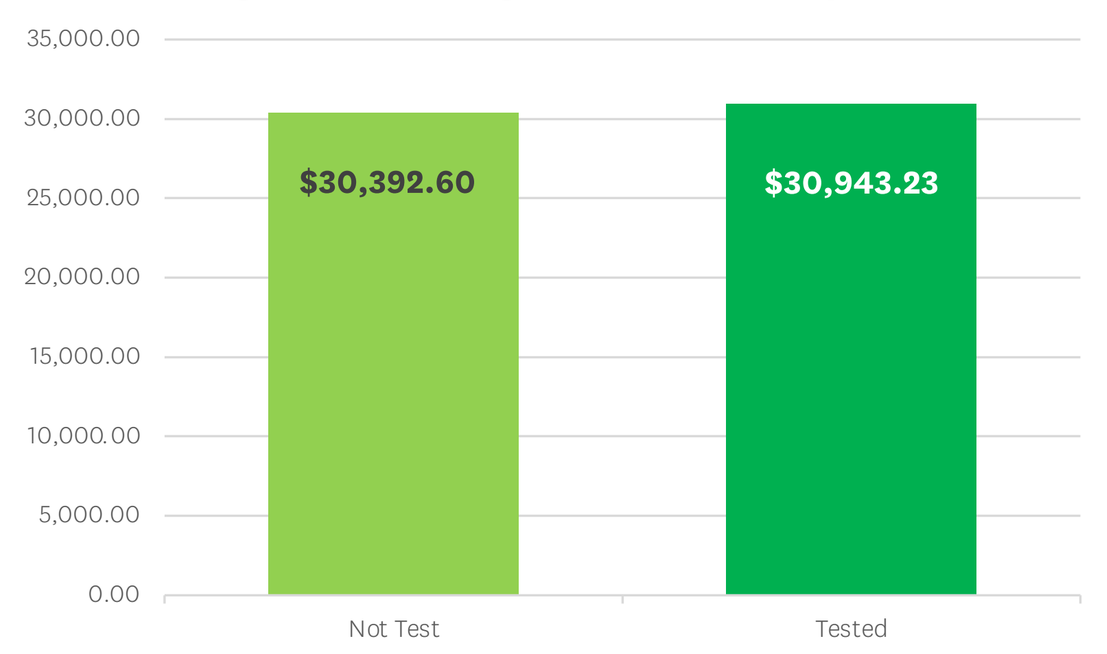
$30,392.60
$30,943.23
Not Test
Tested

Figure 3b. Average Medical PMPM

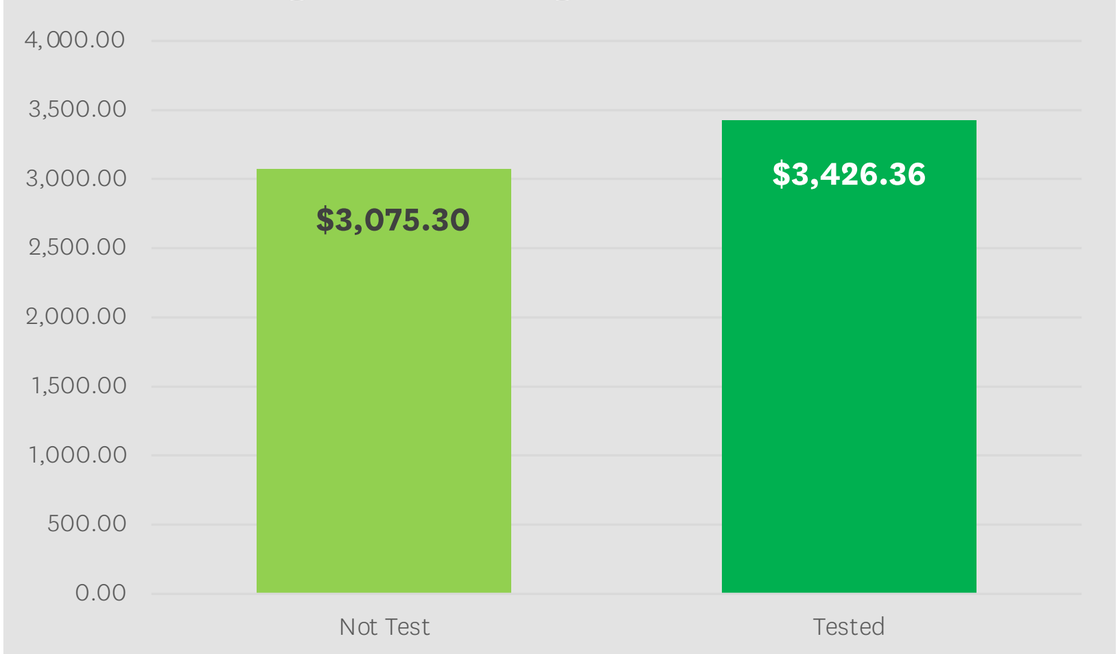
$3,075.30
$3,426.36
Not Test
Tested

Figure 4a. Average Total Pharmacy Spend

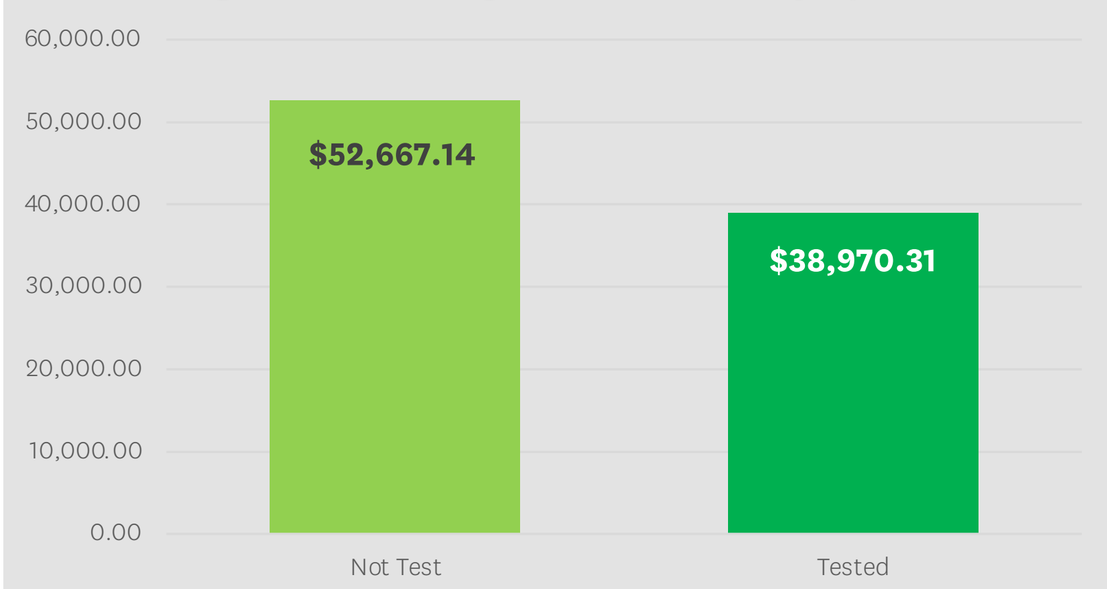
$52,667.14
$38,970.31
Not Test
Tested

Figure 4b. Average Pharmacy PMPM

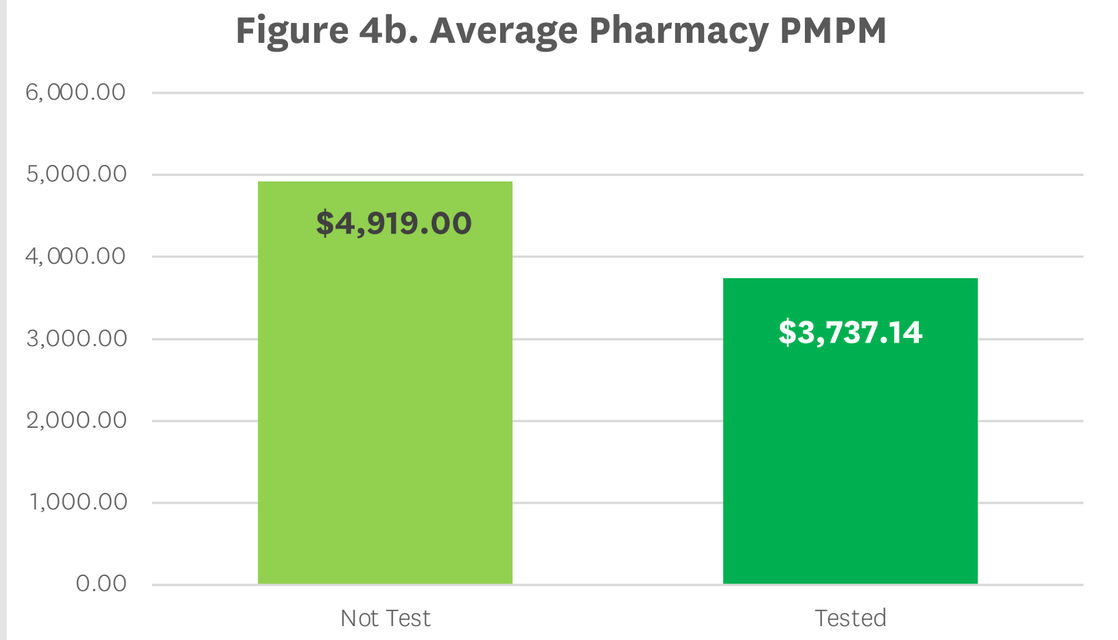
$4,919.00
$3,737.14
Not Test
Tested

Average Total Medical Spend and Average Medical Per Member Per Month (PMPM) (Figure 3a and 3b)

2,379 members identified with the additional inclusion criteria for secondary outcome

After the initial claim of HR+/HER2- mBC therapy, members not tested incurred an average cost of $30,392 and $3,075.30 PMPM. Members tested incurred an average cost of $30,943.23 (p=.73, 95% CI: -3,733.2 – 2,631.9) and $3,426.36 PMPM (p = 0.0525, 95% CI: -705.9 – 3.8268) respectively. These results are not statistically significant.

Average Total Pharmacy Spend and Average Pharmacy PMPM (Figure 4a and 4b)

2,379 members identified with the additional inclusion criteria for secondary outcome:

After the initial claim of HR+/HER2- mBC therapy, members not tested incurred an average cost of $52,667.14 and $4,919.00 PMPM respectively. Members tested incurred an average cost of $38,970.31 (p<0.0001, 95% CI: 7,976.5 – 19,417.2) and $3,737.14 PMPM (p<0.0001), 95% CI: 732.0 – 1631.8) respectively.

CONCLUSIONS

- 48% of the members received an IHC biomarker test prior to receiving a CDK4/6i, AI, Antiestrogen, or ERD. Additional research is needed to determine whether members are being screened prior to treatment for HR+/HER2- mBC.
- Total cost of medical spend and medical PMPM are not statistically significant, while total cost of pharmacy spend and pharmacy PMPM are statistically significant. Further research is required to determine whether there is a difference in cost between the two groups.
- Patient outreach will be important in evaluating whether members are receiving the appropriate biomarker test prior to therapy.
- A clinical program can be created to ensure that patients receive the appropriate biomarker test prior to receiving HR+/HER2- mBC treatment, which can be incorporated into all PA criteria.
- These findings can help inform managed care pharmacy decision making.

ACKNOWLEDGEMENTS

- Thank you to Pfizer, Inc. for funding the AMCP Fdn/Pfizer, Inc. Summer Oncology Internship Program
- Thank you to my phenomenal preceptors for all the time, effort, and patience they gave me:
 - Rodney Gedey, PharmD, Envolve Pharmacy Solutions
 - Ann Wicker, PharmD, BCPS, Pfizer, Inc.
- Special thanks to the following individuals who supported me throughout the capstone research project:
 - Darian Moody, BS, MA, Centene Corp.
 - Dharti Patel, PharmD, MBA, Envolve Pharmacy Solutions
 - Michael Strassner, PharmD, Envolve Pharmacy Solutions
 - Irene Arias, PharmD, MS, BCOP, Pfizer, Inc.
 - Tom Wolfe, PharmD, Pfizer, Inc.
 - Amy Stanford, PharmD, Pfizer, Inc.

REFERENCES S1. SEER Cancer Stat Facts: Female Breast Cancer Subtypes. National Cancer Institute. Bethesda, MD, https://seer.cancer.gov/statfacts/html/breast-subtypes.html 2. National Comprehensive Cancer Network (NCCN). Practice Guidelines in Oncology: Breast Cancer. Version 7.2021. Accessed at https://www.nccn.org/professionals/physician_gls/pdf/breast.pdf.
Poster presented at AMCP Nexus 2021, October 18-21, Denver, CO

© 2021 Envolve. All rights reserved. 6_202110