A Retrospective Analysis of Outpatient Prescription Claim Trends During the COVID-19 Pandemic

Palna Mehta[1], PharmD Candidate, Mark J. Wrobel[2], PharmD, Ying-Fen Swagler[2], PharmD, Brian Gucwa[3], PharmD, Walter J. McClain[3], PharmD/MBA

[1]Rutgers Ernest Mario School of Pharmacy, [2]Independent Health Association, Inc, [3]Pfizer, Inc.

Background

- In response to the COVID-19 pandemic, policies were enacted in New York State (NYS) to ensure patients had access to medications.
- Media reports and peer-reviewed articles fueled speculation about the impact of various drugs/classes on COVID-19, including ACE inhibitors, NSAIDs, and hydroxychloroquine (HCQ).[1-3]
- Fear of opportunistic infections, mental health burden, drug ingredient shortages and manufacturer stoppages associated with social isolation may have influenced prescribing and filling trends.[4,5]
- NYS Governor Andrew Cuomo's lockdown order was signed March 20, 2020 and went into effect March 22, 2020.

Objective

- To find outpatient prescription drugs/classes with changes in claim trends at a medium-sized health maintenance organization (HMO) in western NYS after Gov. Cuomo's COVID-19 emergency stay-at-home order, focusing on drugs/classes discussed in the media.

Methods

- Retrospective study of outpatient pharmacy claims at a 350,000-life HMO covering 8 counties in western NYS.
- Claims information from January 1 to May 31 in 2019 and 2020 included patient age, line of business, zip code, drug name, generic product identifier (GPI) name and number, claim date, and quantity filled.
- Drug classes were sorted by first 4-digit GPI into month and year of fill and normalized by member months.
- Cumulative list of normalized differences and percentage changes between 2019 and 2020 for all classes were compiled separately for overall January through May data and individual months.
- Data were plotted as a bell curve; any value greater than 1 standard deviation from the monthly mean was investigated to see if specific drugs drove the overall class trend.
- Drugs/classes mentioned by the media or scientific journals were also examined if they were not identified in the previous step.[1-5]
- Normalized script counts of the drugs and drug classes of interest were analyzed weekly between 2018/2019 and 2019/2020 to see if there were any notable trends in weekly utilization.
- Data were analyzed using Microsoft Excel and an interrupted time series was performed using STATA.

Results

% changes in normalized script count from 2019 to 2020:

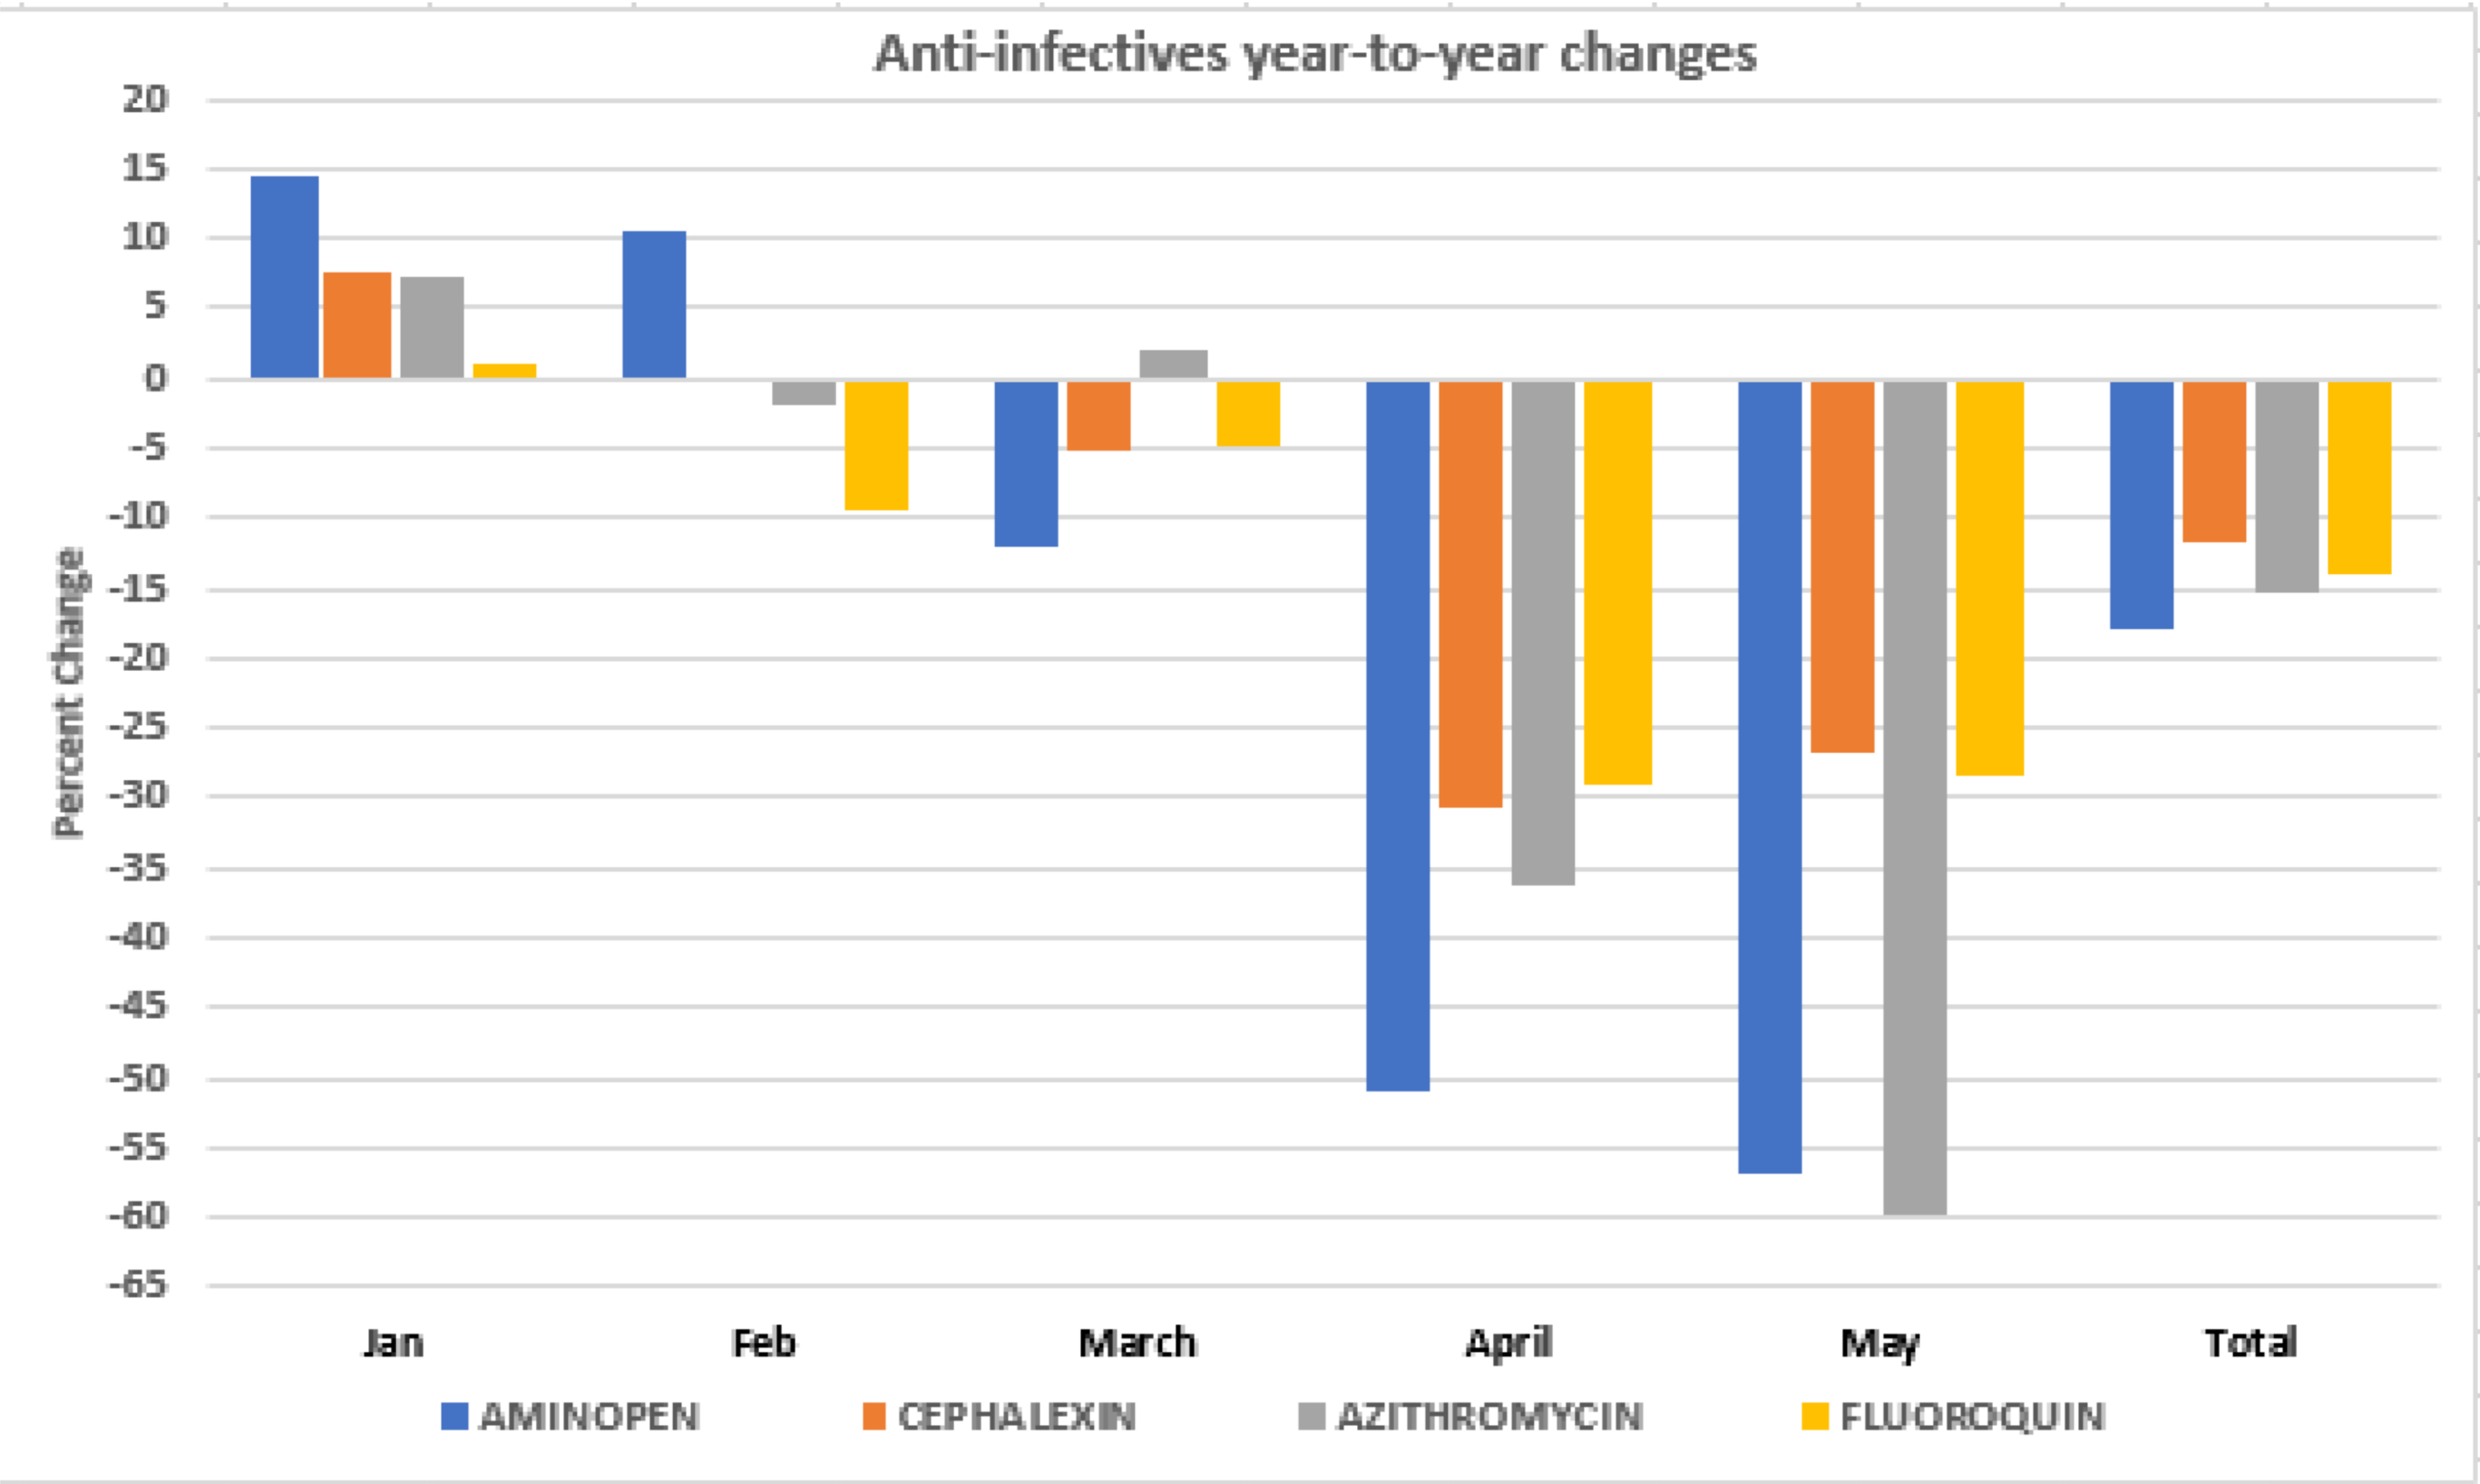

Anti-infectives year-to-year changes
Percent Change
20
15
10
5
0
-5
-10
-15
-20
-25
-30
-35
-40
-45
-50
-55
-60
-65
Jan
Feb
March
April
May
Total
AMINOPEN
CEPHALEXIN
AZITHROMYCIN
FLUOROQUIN

- Year-to-year relative prescription count decreases >25% in major anti-infective classes in the two months following Gov. Cuomo's stay-at-home orders.

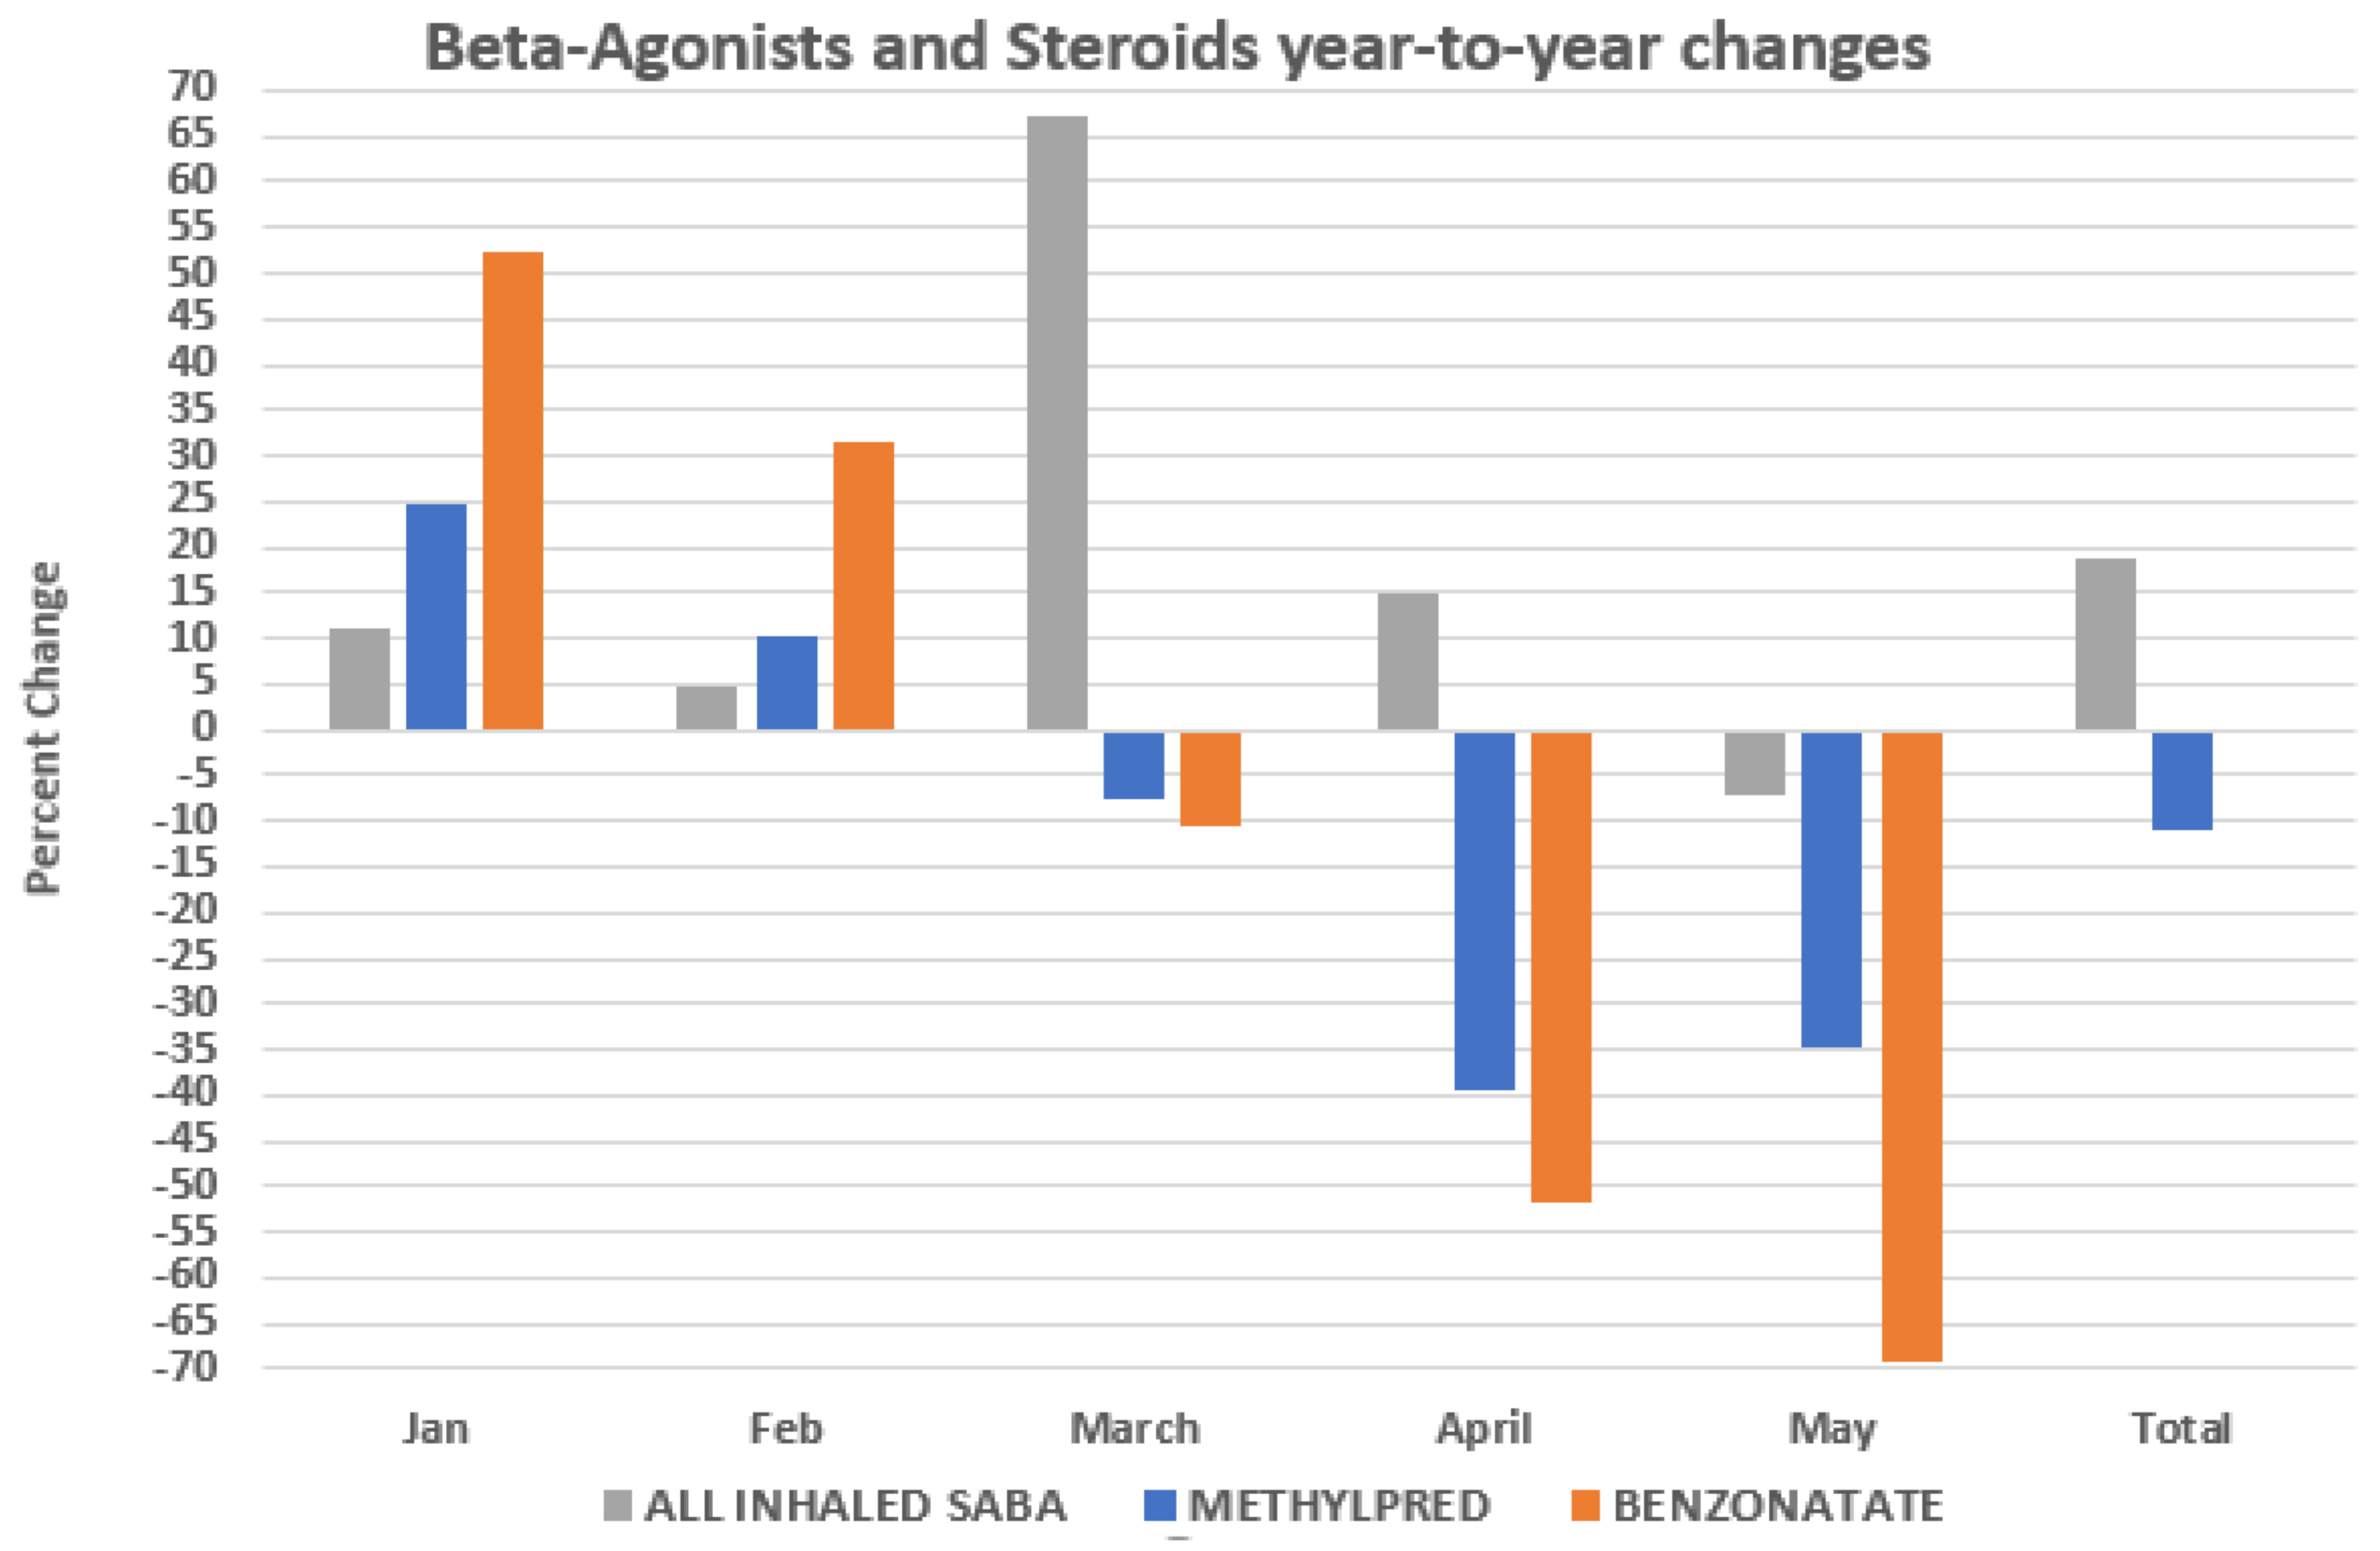

Beta-Agonists and Steroids year-to-year changes
Percent Change
70
65
60
55
50
45
40
35
30
25
20
15
10
5
0
-5
-10
-15
-20
-25
-30
-35
-40
-45
-50
-55
-60
-65
-70
Jan
Feb
March
April
May
Total
ALL INHALED SABA
METHYLPRED
BENZONATATE

- Similar large declines in relative prescription count were seen with steroids and benzonatate, a medication for acute cough.
- Short-acting beta-agonist (SABA) fills spiked in March and decreased 7.00% in May from 2019 to 2020; coincided with concerns about a possible COVID-19-related albuterol shortage.

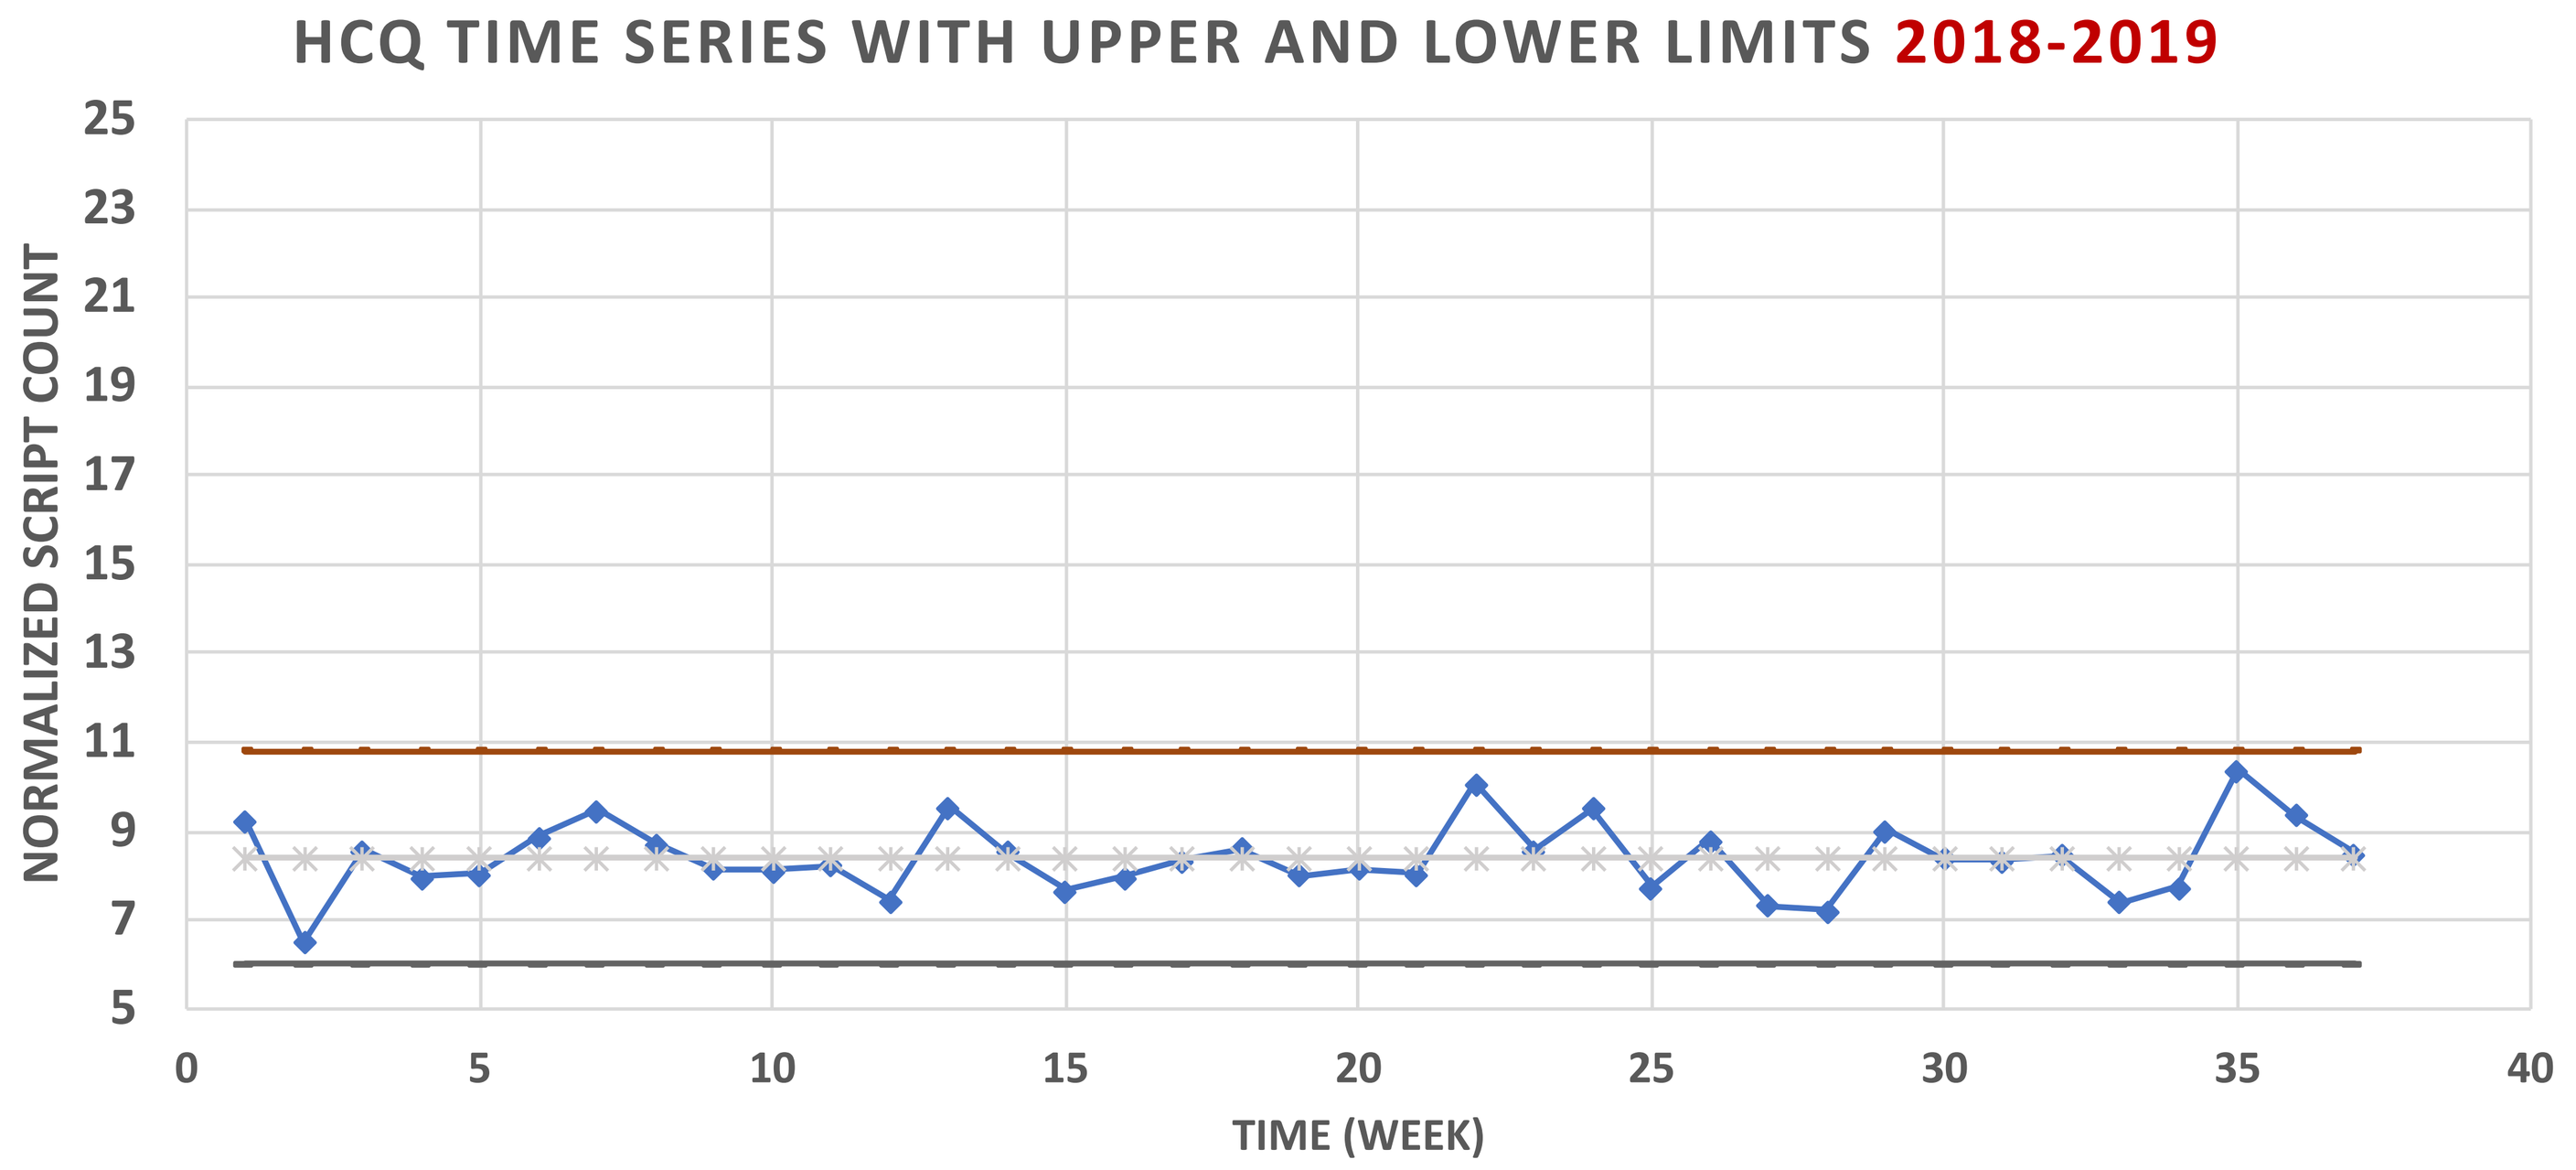

HCQ TIME SERIES WITH UPPER AND LOWER LIMITS 2018-2019
NORMALIZED SCRIPT COUNT
25
23
21
19
17
15
13
11
9
7
5
0
5
10
15
20
25
30
35
40
TIME (WEEK)

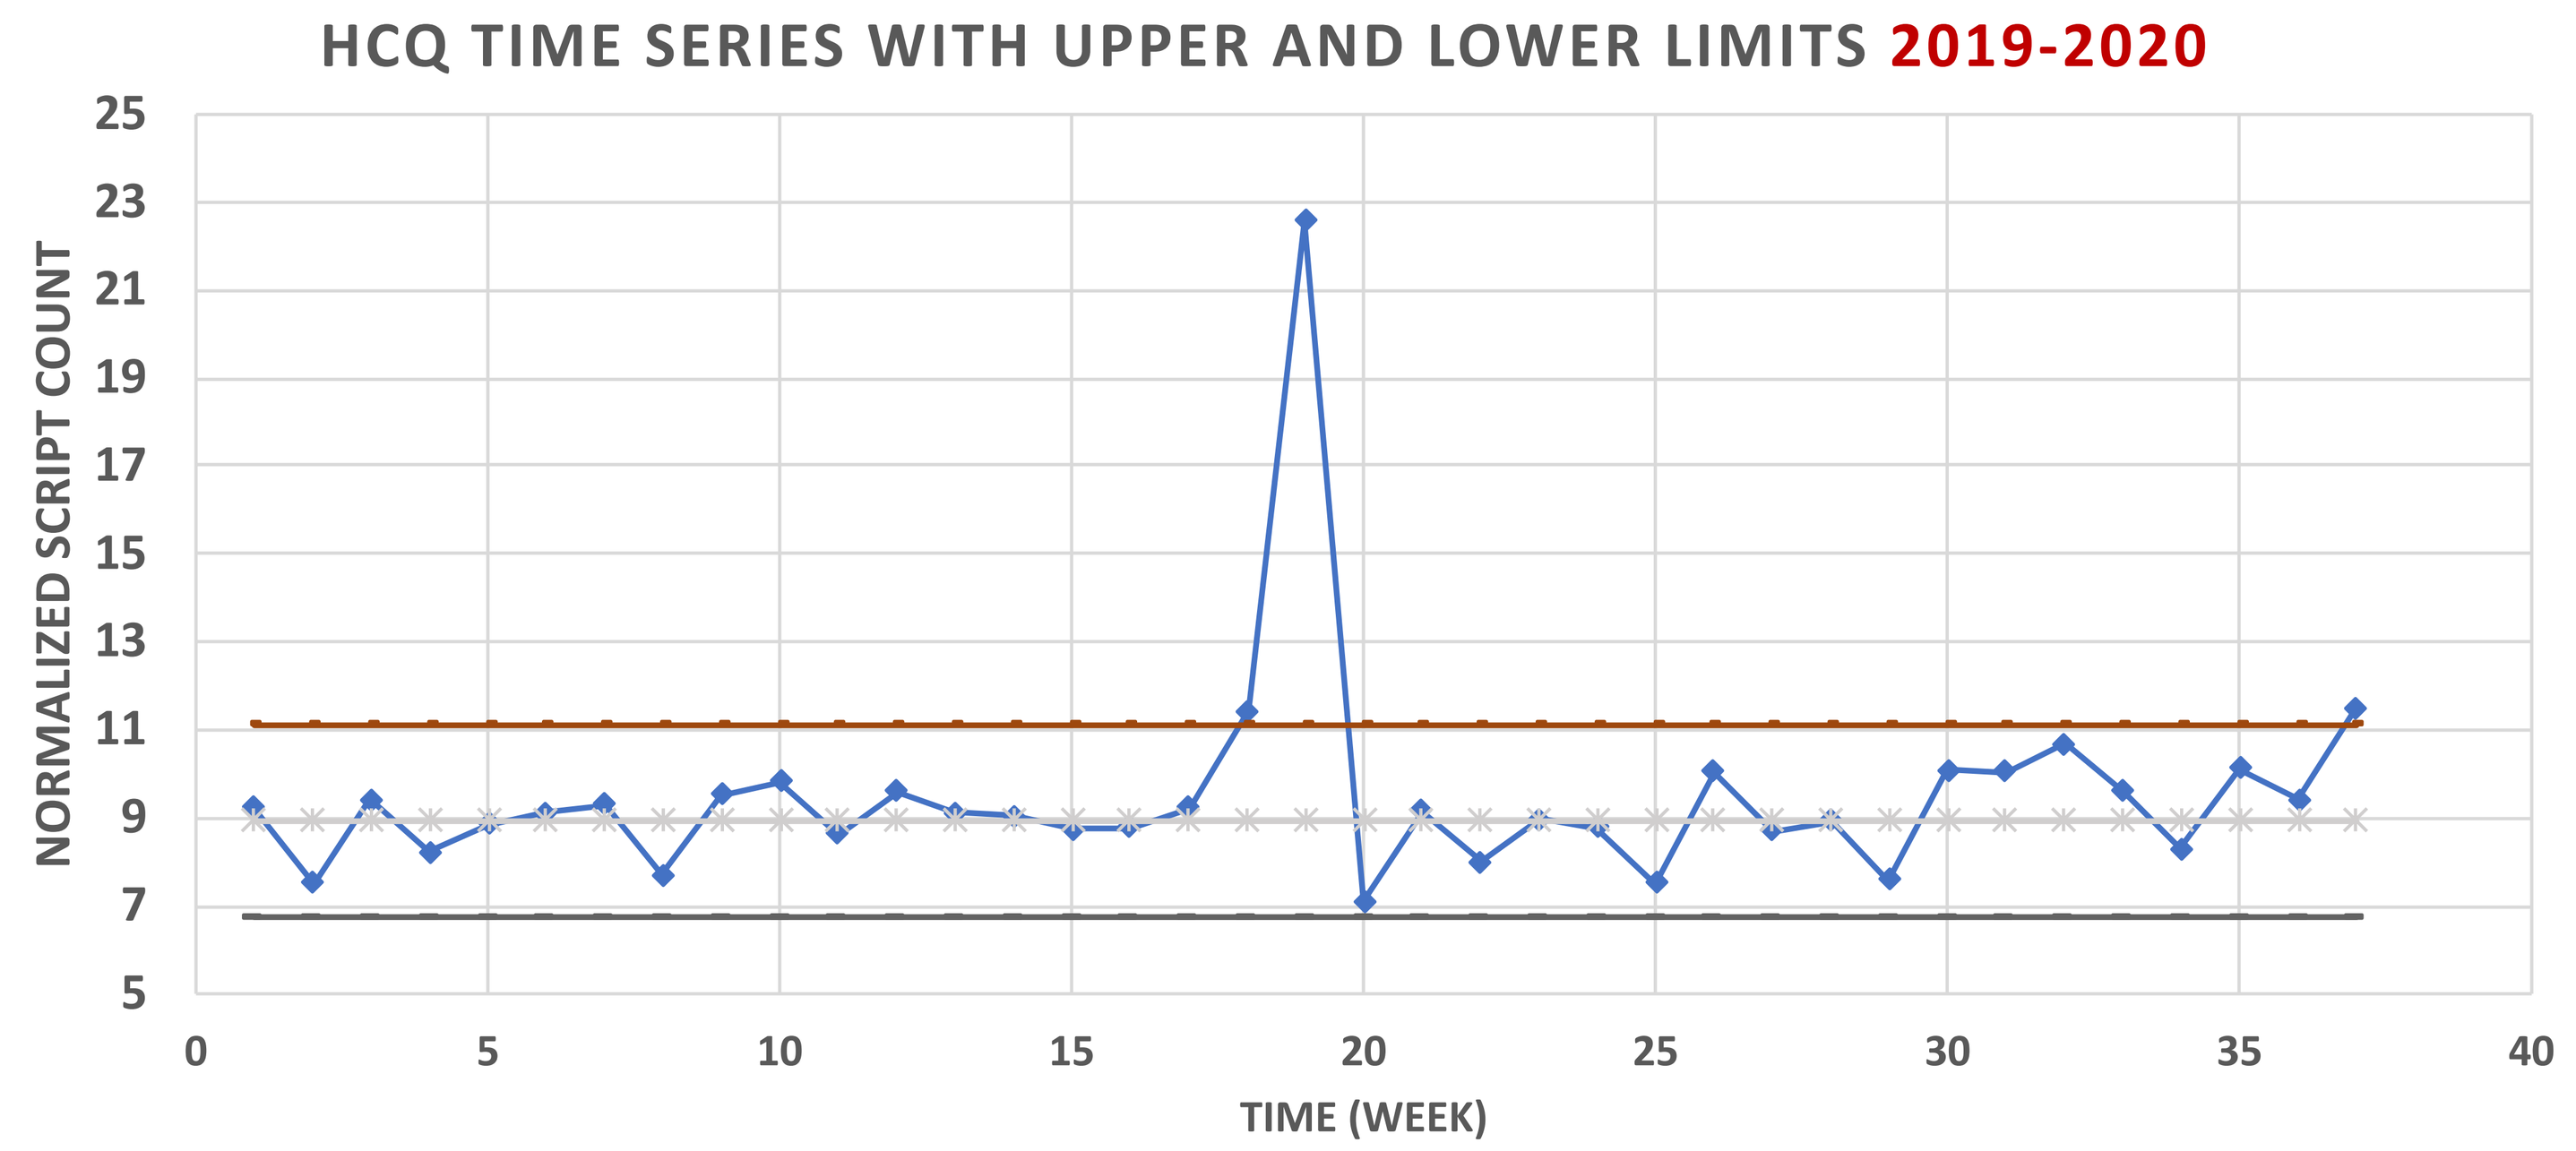

HCQ TIME SERIES WITH UPPER AND LOWER LIMITS 2019-2020
NORMALIZED SCRIPT COUNT
25
23
21
19
17
15
13
11
9
7
5
0
5
10
15
20
25
30
35
40
TIME (WEEK)

- Reference week 1 represents week 46 of the first year and the numbering is continuous through week 30 of the following year.
- March 2020 saw a 50.7% relative increase in HCQ fills compared to March 2019.
- A 2020 weekly run chart showed claim trends within a normal range of values until week 19 (week starting March 15, 2020).
- In the following week, on March 23, 2020, Gov. Cuomo signed an executive order limiting HCQ dispensing to FDA-approved indications only.
- The HMO implemented an indication-based prior authorization for HCQ on April 1, 2020 to enforce the executive order.

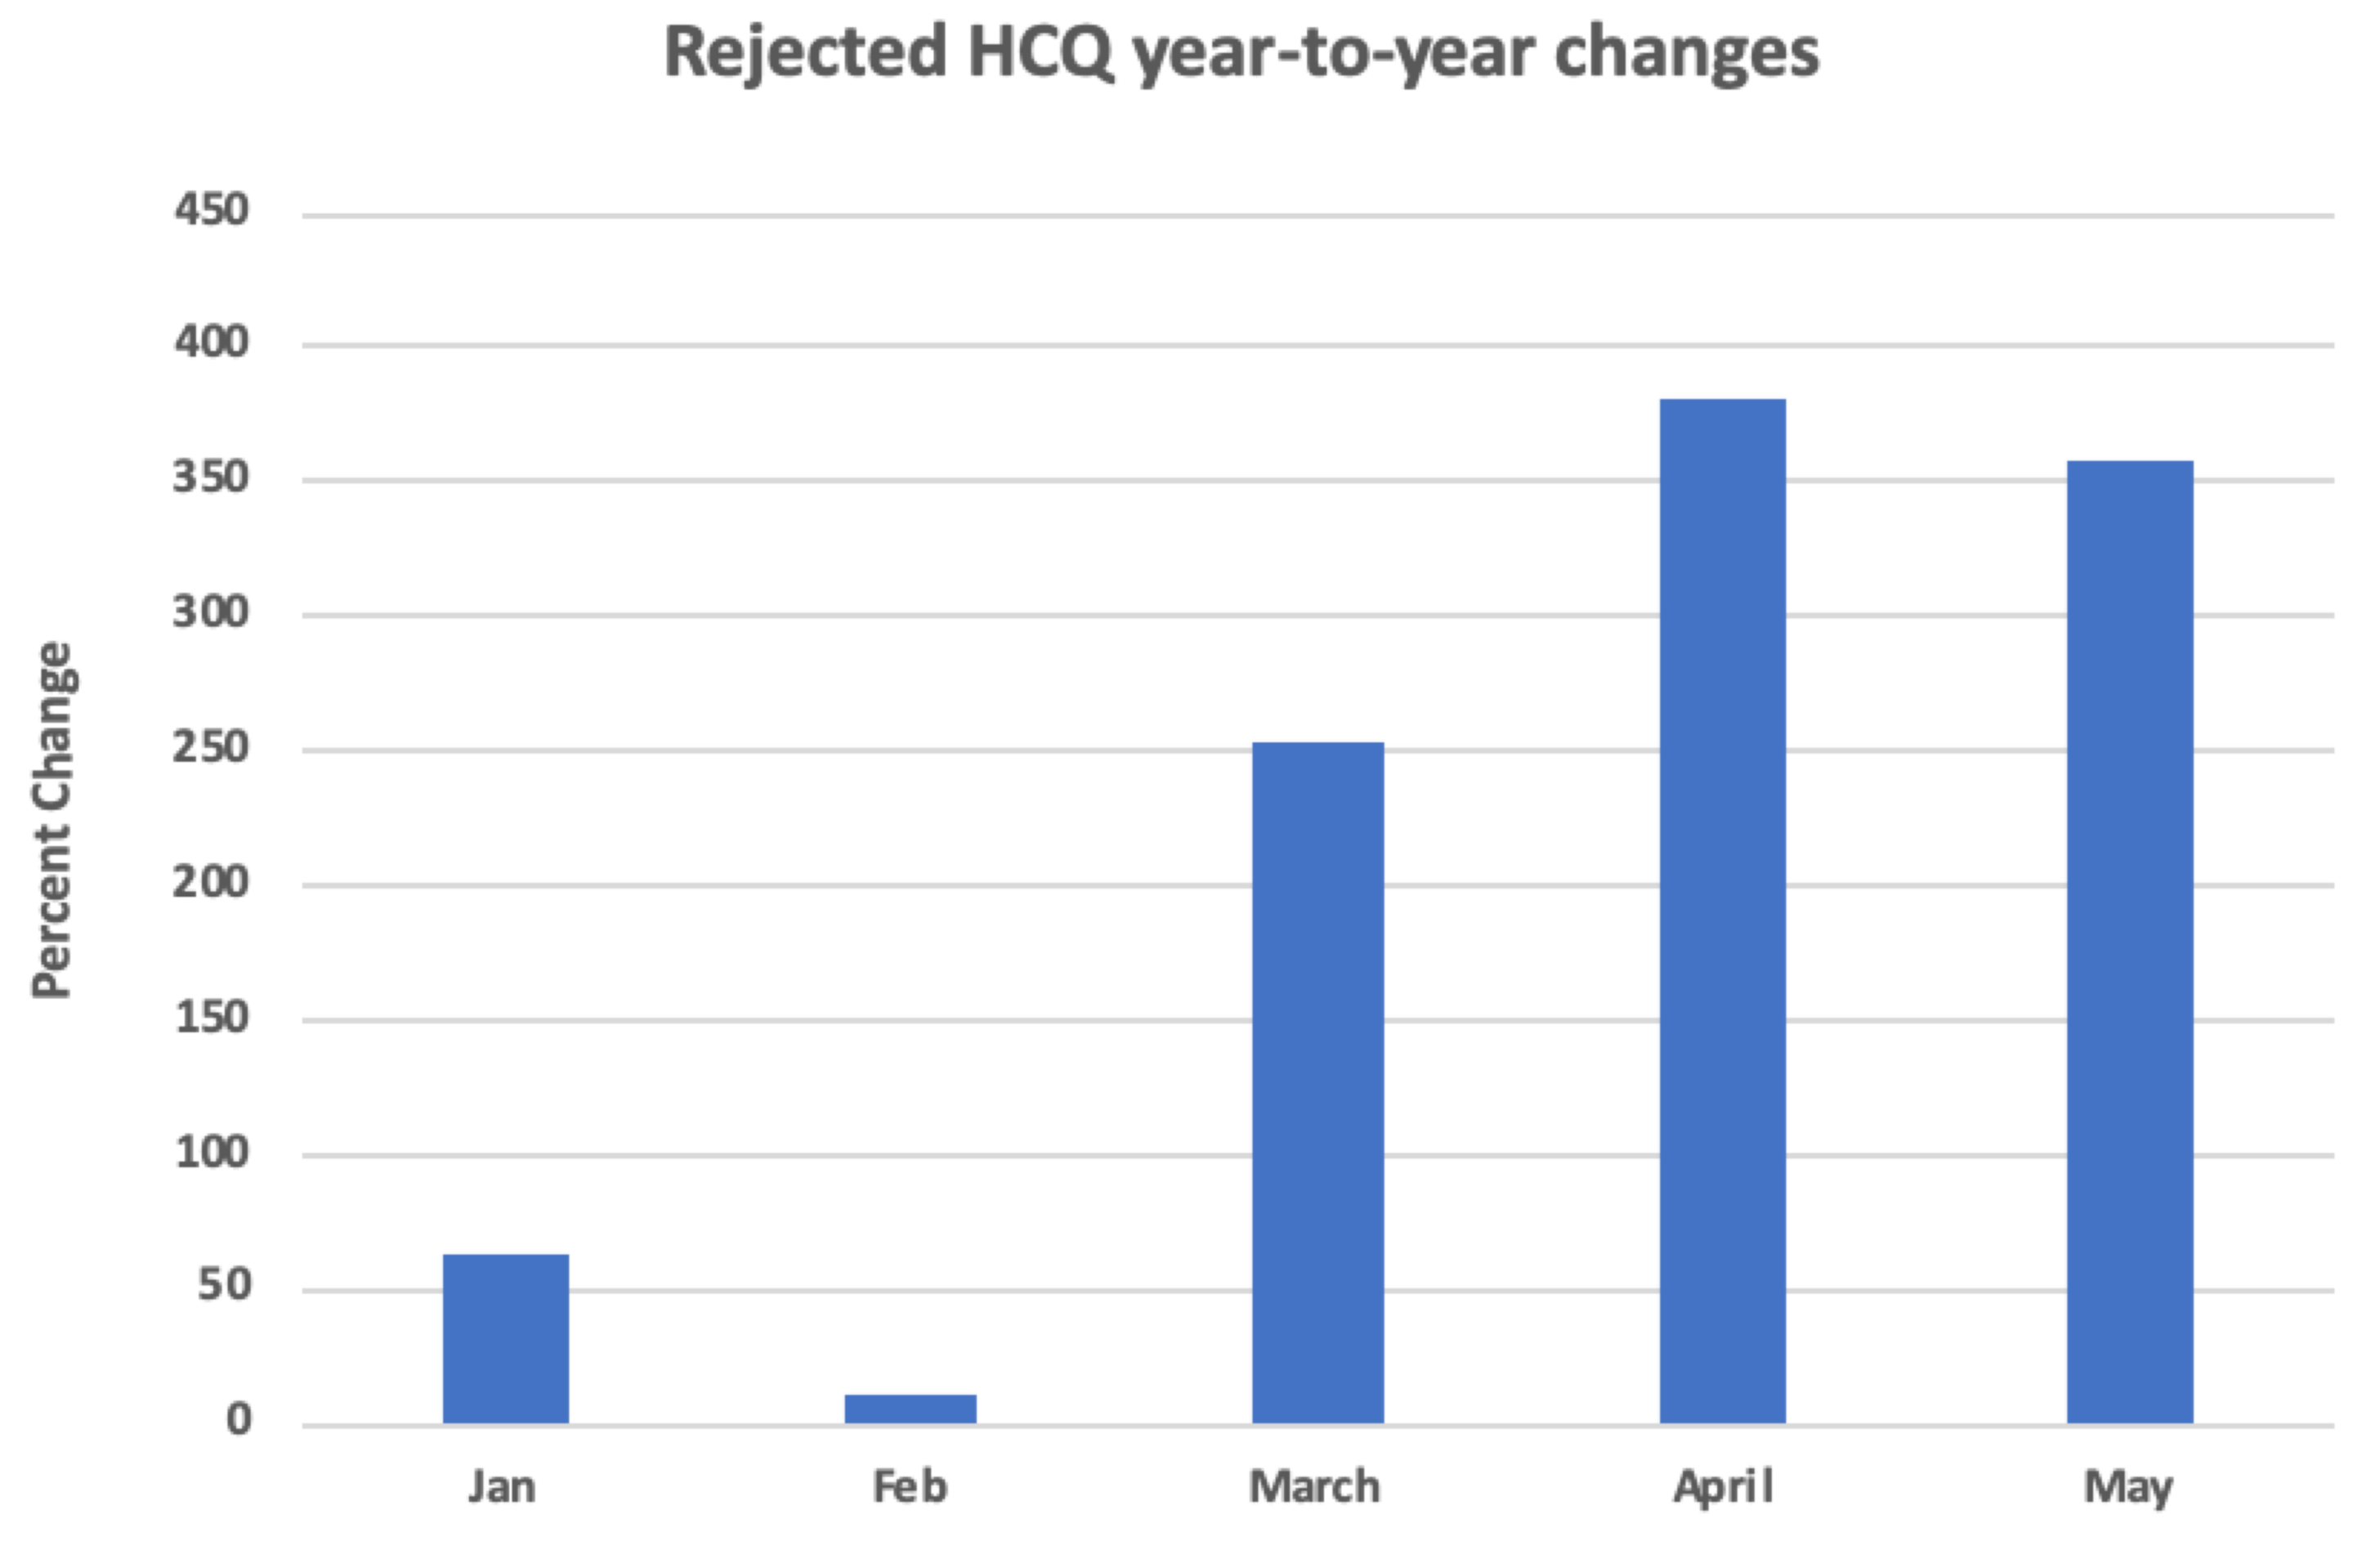

Rejected HCQ year-to-year changes
Percent Change
450
400
350
300
250
200
150
100
50
0
Jan
Feb
March
April
May

- Year-to-year rejected HCQ claims increased 254% in March 2020, 382% in April 2020, and 359% in May 2020.
- Compared with the prior 16 weeks, there was a statistically significant increase of 6.4 rejected HCQ claims per week for the 21 weeks beginning March 2020 ($p<0.001$, 95% CI 3.0-9.9).
- Despite mounting evidence throughout the 21 weeks against HCQ's efficacy for COVID-19,[6] claim rejections remained steady throughout the study period.

Limitations

- Observed trends cannot establish causation without further statistical analysis and data on diagnostic codes or intended use.
- Data were normalized but no mathematical method can account for other factors that could explain fluctuations in trends.

Conclusions

- HCQ claim increases and continued rejections coincided with media coverage of COVID-19.
- Decreases in claim trends could be explained by fewer patients seeking medical attention for mild infections and symptoms.
- If decreases in anti-infective, steroid, and cough and cold medication were caused in part by decreased social interaction due to stay-at-home orders, there may be a benefit to socially distancing (working from home, remote learning) during illness.

References

1. Fang L et al. *Lancet Resp Med* 2020;8:e21.
2. Gautret P et al. *Int J Antimicrob Agents* 2020;105949.
3. Gao J et al. *Biosci Trends* 2020;14(1):72-3.
4. Huang Y and Zhao N. *Psychiatry Res* 2020;288:112954.
5. Jacobs A. Doctors Heavily Overprescribed Antibiotics Early in the Pandemic. *The New York Times*. June 9th 2020. Accessed August 5th 2020.
6. Boulware DR et al. *N Engl J Med* 2020;383:517-25

Poster presented at Academy of Managed Care Pharmacy Nexus 2020
Virtual, October 19 - October 23, 2020