AMCP Nexus 2020

The Emergence of Therapeutic Oncology Biosimilars: Payer's Perspective

Aisha Fowler, PharmD Candidate, Rebecca Borgert, PharmD, BCOP, Michael Polson, PharmD, Kenneth Lawson, PhD , Jim Rebello, PharmD, Haita Makanji, PharmD

Special Acknowledgments: Sarah Hanson, MD, PhD, MBA, Bethany Boyd, MS, PhD, Ahmed Shelbaya, MD, MSc, MPH

Magellan Rx Management

Background

- The total cost of cancer care in 2020 were estimated to be $207 billion, representing a 66% increase since 2010.[1]
- Over the last two decades, the use of biologic drugs for the treatment of cancer has expanded rapidly.
- In particular, bevacizumab (Avastin®), rituximab (Rituxan®), and trastuzumab (Herceptin®) accounted for the highest cancer drug spend from 2011 to 2016, totaling $8.6 billion in the United States (U.S.).[2]
- Since 2019, nine FDA-approved biosimilar versions have entered the marketplace including:
 - bevacizumab biosimilars: Mvasi®, Zirabev™
 - trastuzumab biosimilars: Kanjinti™, Ogivri™, Ontruzant®, Herzuma®, Trazimera™
 - rituximab biosimilars: Truxima®, Ruxience™

Objectives

- Characterize the early utilization of therapeutic oncology biosimilars for bevacizumab, trastuzumab, and rituximab in commercial and Medicare lines of business
- Estimate the budgetary impact of increased biosimilar utilization for oncology treatment from the payer's perspective

Methods

Study Design

- Retrospective, cross-sectional analysis of cancer patients who initiated the selected biologics or biosimilars for oncology treatment between 6/16/2019 and 6/16/2020 using prior authorization (PA) and administrative medical claims data

Inclusion Criteria

- Commercial and Medicare members with ≥1 PA + ICD-10 diagnosis code for cancer
- Adjudicated medical claims + ICD-10 diagnosis codes for cancer

Exclusion Criteria

- Data from Medicaid plans
- PA approvals or administrative claims for:
 - Subcutaneous formulations
 - Ophthalmic injections
 - ICD-10 codes for non-oncology use
 - HCPCS code J9999

Data Sources

- Magellan Rx Management (MRx) PA and medical claims databases
- 11 million MRx Medical Pharmacy lives
- 500,000 Managed Medicare lives (regional health plans)

Characterizing Trends & Utilization

- Formulary decisions by health plans, as identified in the PA requests, were categorized based on their biosimilar coverage polices as either:
 - ***Biosimilar preferred*** – if the plan required a step-through (ST) biosimilar before gaining access to the innovator product
 - ***On par*** – if the plan did not prioritize either the biosimilar or innovator biologic
 - ***Non-preferred*** – if the plan required the innovator product before gaining access to the biosimilar
- In addition, patterns of ***switching*** from the innovator biologic to an FDA-approved biosimilar were summarized based on the *site of service.* Switching was identified if:
 - Same patient had a PA for > 1 medication with the same active ingredient; and
 - Innovator biologic was approved 30 days to 210 days before biosimilar

Statistical analysis

- PA approvals were categorized based on active drug and biosimilar vs. innovator biologic.
- Descriptive statistics were generated to describe categorical variables (count and percentage).

Budget Impact

- A budget impact model was conducted using administrative medical claims over a 1-year time horizon. Costs were estimated using the ***total allowed amount*** - which was the maximum amount a plan paid for the biologic, and reported as the average cost per claim.
- Market shares were based on the observed total allowed paid.
- The model was also run from the payer perspective and included commercial and Medicare health plans from the Magellan Rx Management administrative medical claims database.

Biosimilars for oncology treatment show increased uptake and potentially sizable savings for health plans.

Results

Figure 1: Formulary coverage by plans

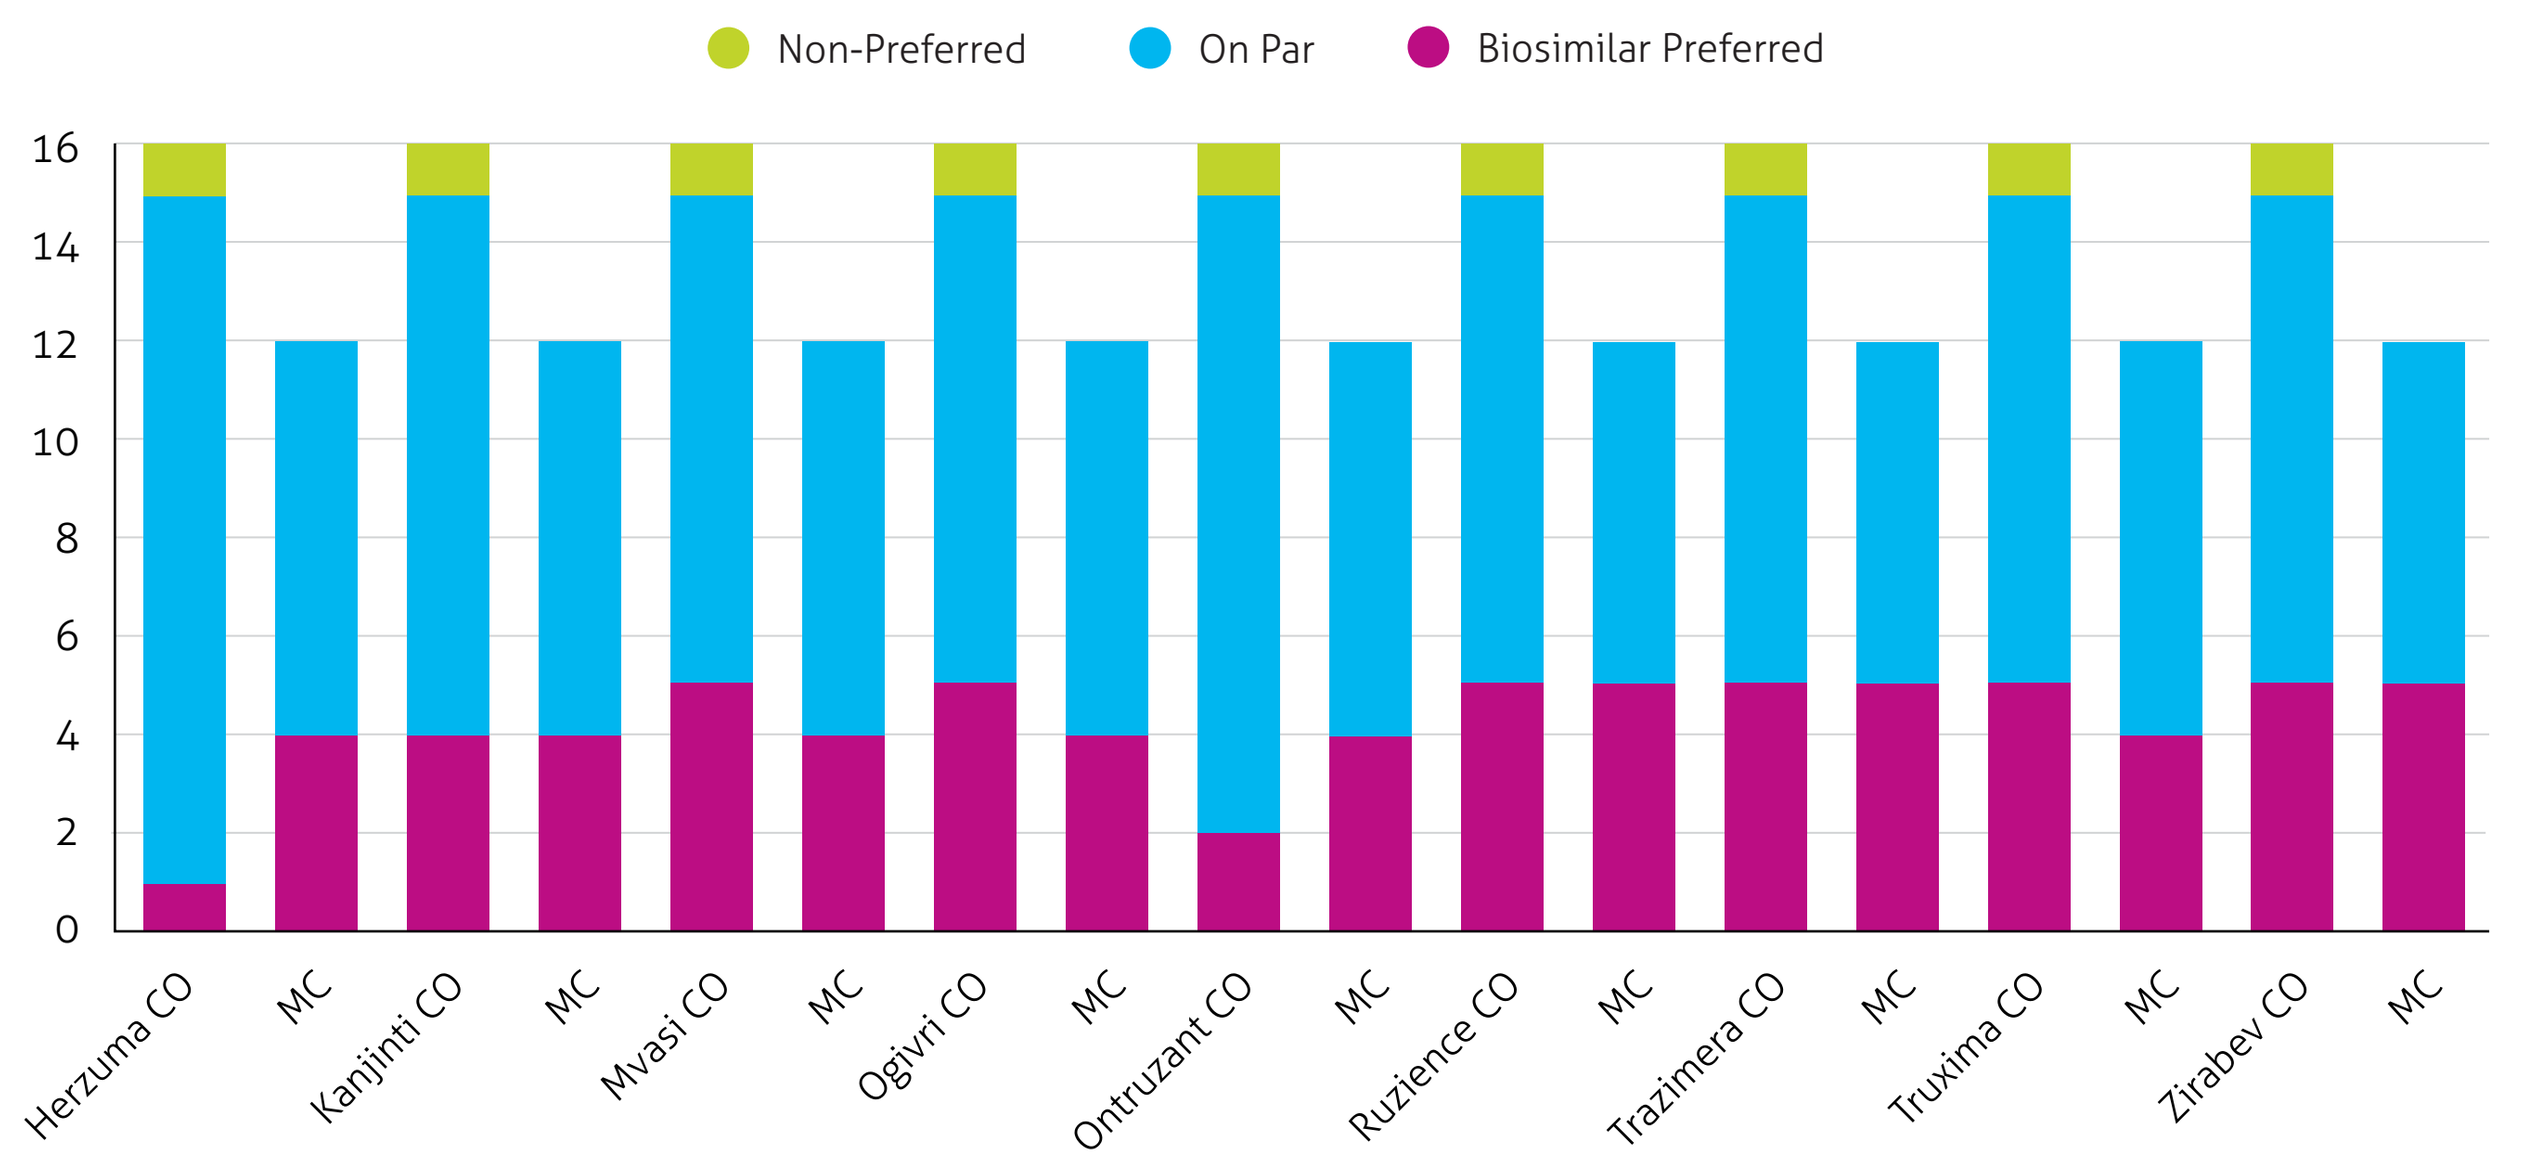

Non-Preferred
On Par
Biosimilar Preferred
16
14
12
10
8
6
4
2
0
Herzuma CO
MC
Kanjinti CO
MC
Mvasi CO
MC
Ogivri CO
MC
Ontruzant CO
MC
Ruzience CO
MC
Trazimera CO
MC
Truxima CO
MC
Zirabev CO
MC

CO - commercial plans (n = 16)
MC - Medicare Advantage plans (n = 12)

Figure 2: Market share based on PA approvals between 6/16/2019 and 6/16/2020

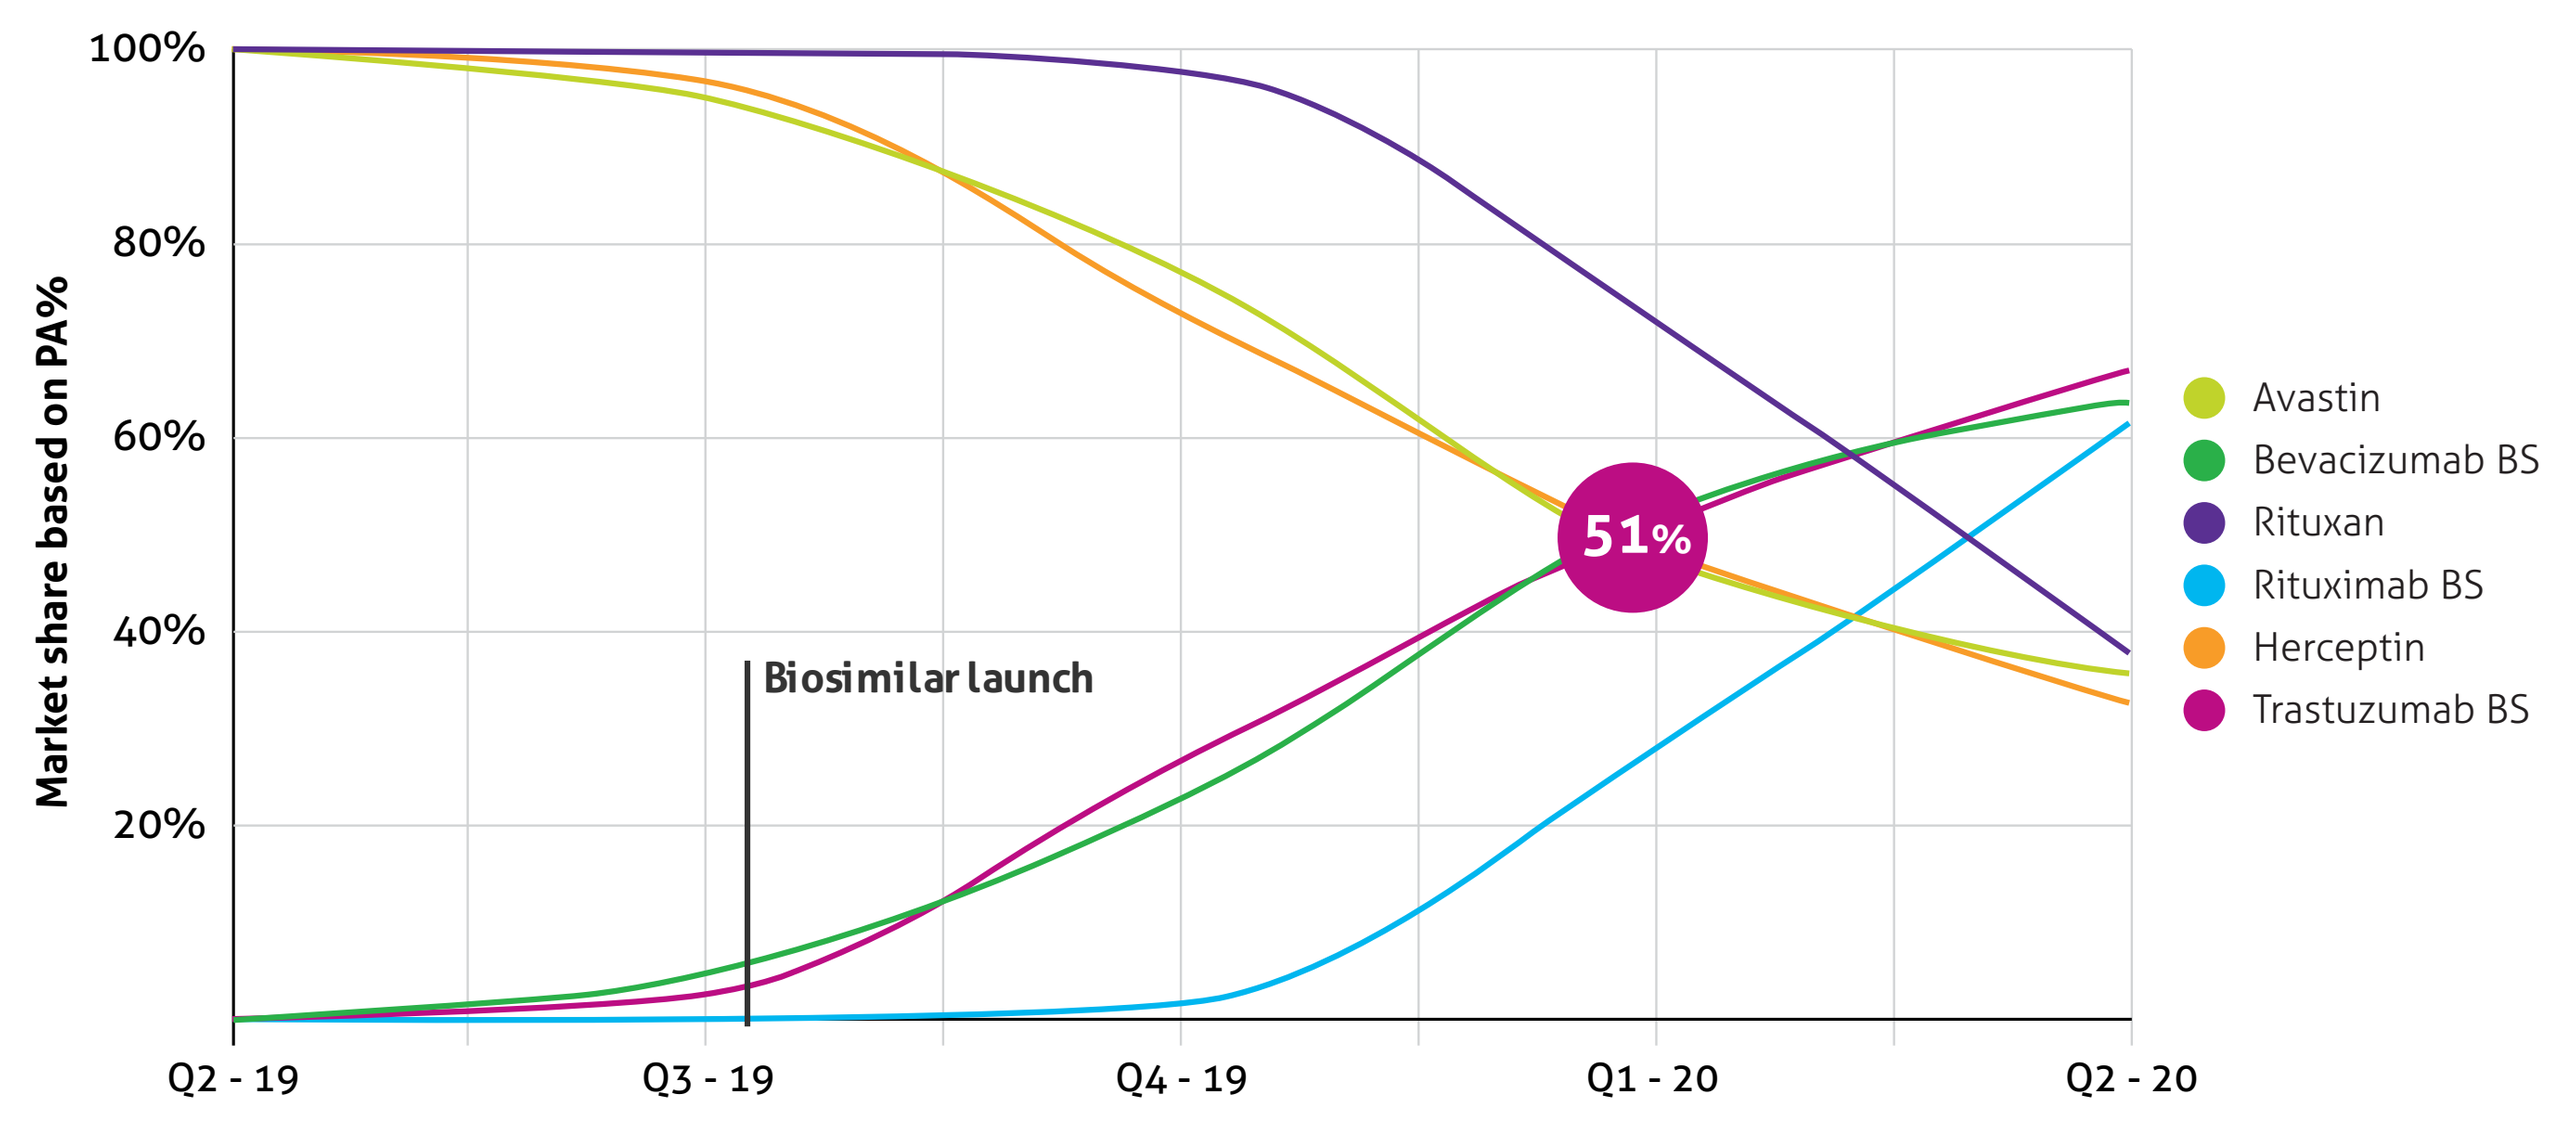

Market share based on PA%
100%
80%
60%
40%
20%
Biosimilar launch
51%
Q2 - 19
Q3 - 19
Q4 - 19
Q1 - 20
Q2 - 20
Avastin
Bevacizumab BS
Rituxan
Rituximab BS
Herceptin
Trastuzumab BS

Total PA approvals= 7,591; Biosimilar PA approvals = 2,217; Originator Biologic PA approvals = 5,374

Figure 3: Independent switches from the innovator biologic to biosimilar

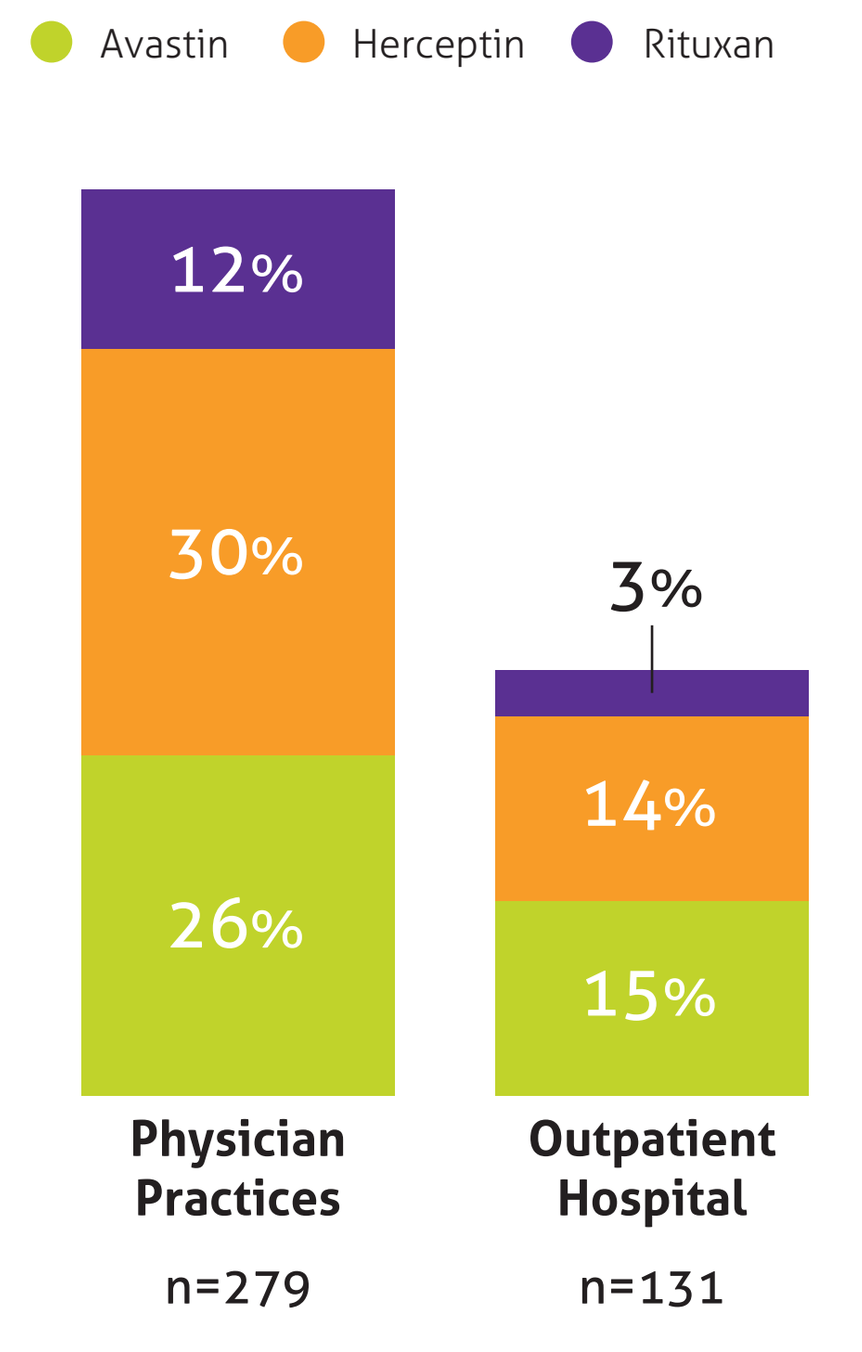

Avastin
Herceptin
Rituxan
12%
30%
26%
3%
14%
15%
Physician Practices
n=279
Outpatient Hospital
n=131

Originator Biologic PA approvals = 5,374
Biosimilar Switches identified = 410

Table 1: Budget impact over 1 year—Medicare and commercial perspectives

	Observed			Estimated				Budget Impact	
	Total Allowed Amt Paid	Number of Claims	Market Mix	Allowed Amt Paid per Claim	Number of Claims	Market Mix	Total Allowed Amt Paid	Difference	%
Bevacizumab Biosimilars	$11,329,205	2827	7%	$4,008	20076	50%	$80,454,590		
Avastin	$226,645,186	37325	93%	$6,072	20076	50%	$121,905,660	($35,614,141)	-15%
Trastuzumab Biosimilars	$11,338,653	2328	5%	$4,871	25511	50%	$124,252,741		
Herceptin	$261,684,455	48694	95%	$5,374	25511	50%	$137,097,633	($11,672,735)	-4%
Rituximab Biosimilars	$1,795,909	335	2%	$5,361	9275	50%	$49,722,565		
Rituxan	$138,262,974	18215	98%	$7,591	9275	50%	$70,402,914	($19,933,404)	-14%
TOTAL BUDGET	$651,056,384						$583,836,104	($67,220,280)	

Discussion

- A total of 7,591 PA approvals met the inclusion criteria, including 5,374 for innovator biologics and 2,217 for biosimilars.
- Based on 252 medical formulary decisions across the 16 commercial and 12 Medicare Advantage plans, an average total allowed of 30% granted biosimilar preferred coverage; 4% non-preferred coverage; and 66% on par coverage.
- Based on PA approvals, the uptake of therapeutic oncology biosimilars grew steadily since launch, and biosimilar utilization surpassed the utilization of innovator biologics during the 1st quarter of 2020.
- There are currently no FDA-approved biosimilars with an interchangeable designation. However, of 5,374 PA approvals for innovator biologics, 410 (8%) were independently switching from the innovator biologic to the biosimilar during the middle of their course of therapy.
 - Switching occurred more frequently in physician practices (68%) than outpatient hospital departments (32%)
- A budget impact model was calculated from the average amount paid per claim using medical administrative claims from the MRx database (n= 109,724). Based on the actual observed market share achieved in Q1-2020 (~51%), the model assumed a transition to a 50% biosimilar market share over one year.
 - The budget impact model yielded an estimated $35,614,141, $11,672,735, and $19,933,404 lower total allowed amount paid for bevacizumab, trastuzumab, and rituximab, respectively.

Conclusion

- Payers equipped with proactive utilization management strategies for oncology biosimilars were able to capitalize on early utilization shifts to the less expensive biosimilar products.
 - Specifically, step therapy requirements increased use of biosimilars, and were implemented successfully with minimal disruption.
- Data also indicates that some providers are proactively switching patients to oncology biosimilars even without biosimilar preferred coverage.
- Increased biosimilar utilization of 50% market share will have a sizeable budget impact on US commercial and Medicare health plans.
- Longer follow-up and corresponding claims analysis may provide further insights on the true uptake in biosimilar use and cost savings being achieved.

Limitations

- Prior authorization determinations may not be fully reflective of biosimilar use, as patients may have received alternate therapy, never received the biosimilar, or received the reference product through manufacturer assistance programs for clinically appropriate reasons that cannot be discerned from the data without corresponding claims.
- Due to variable rebate arrangements in place for different payers, true savings are unable to be determined yet.

References

1. Mariotto AB, Robin Yabroff K, Shao Y, Feuer EJ, Brown ML. Projections of the cost of cancer care in the United States: 2010–2020. Journal of the National Cancer Institute. 2011 Jan 19;103(2):117-28.
2. Hong SJ, Li ED, Matusiak LM ,et al. Spending on antineoplastic agents in the United States, 2011 to 2016 J Oncol Practice 2018;14: e683-e691.
3. Magellan Rx internal data, 2019-2020.

Disclosures

- Thank you to Pfizer, Inc. for funding the AMCP Foundation Managed Care Oncology Summer Internship Program.

Aisha Fowler
aishafowler@utexas.edu

More clinical research from **Magellan Rx**→