Aligning Drug Development, Regulatory Approval, and Managed Care Pharmacy: A Cross-Sectional Study Addressing Evidence Gaps in FDA Expedited Approvals

Riana Roberts[1], PharmD Candidate; John Spain[2], MA, PharmD, BCPS; Annesha White[3], PharmD, MS, PhD; Paula Eichenbrenner[4], MBA, CAE; Terry Richardson[5], PharmD, BCACP; Vyishali Dharbhamalla[6], PharmD; Claire Gorey[7], PhD Candidate

ETSU Bill Gatton College of Pharmacy[1], Pfizer, Inc.[2], University of North Texas System College of Pharmacy[3], AMCP[5] & AMCP Foundation[4,6], Xcenda[7]

Background

- In recent years, there has been an increase and expanded use of expedited approval programs.[1]
- There are four pathways in which the FDA can allow for earlier approval of drugs: (1) fast track designation, (2) breakthrough designation, (3) accelerated approval, and (4) priority review.
- In the process of approving drugs via expedited pathways, the payers' perspective may differ from the FDA's approval criteria.[2,3]
- With both the benefits and substantial risks of medications approved before confirmatory trials, it is essential to address these concerns as well as to highlight solutions and barriers regarding evidence for new drugs.[4]
- This study illuminates stakeholder concerns across formulary decision-making; health care resource utilization; real-world evidence; drug development; and patient access.
- Several questions were repeated from the AMCPF Trends in Healthcare Survey (2018) to assess changes in perspectives on FDA expedited approvals.[5]

Objectives

1. To identify evidence gaps to provide guidance on improving the expedited approval process.
2. To ascertain treatment outcomes valued by payers, so that links from surrogate endpoints to meaningful outcomes are better understood by all stakeholders in drug development, regulatory approval, and formulary decision-making.
3. To assess how payers' perspectives about accelerated drug approvals have shifted in the past three years.

Limitations

- The respondents' level of expertise and awareness cannot be verified or proven. Caution is advised when generalizing the survey results.
- Measures were taken to ensure an even distribution in the demographics. However, the results may be skewed towards the views of specific groups due to different response rates.

Methods

- This cross-sectional study consisted of a web-based survey with 22 multiple choice and open-ended questions.
- The survey instrument was circulated to pilot respondents (n=8) to assess content and face validity.
- AMCP's internal CRM database was used to filter for groups of key health care decision makers and other AMCP Foundation stakeholders.

Employer Type	Job Function	Expertise
• Payers, pharmacy benefit managers, specialty pharmacy	• Pharmacy director, formulary management, operations, contracting	• Formulary and/or commercial/ex-change plan management

- Inferential and descriptive statistics were used to analyze the data.
- Conducted narrative literature reviews on the topic of FDA expedited approvals.
- The first 200 respondents who fully completed the survey are scheduled to receive a $10 Amazon e-gift card. The target for complete responses was 200.
- Survey was fielded from September 22-October 4, 2021.

Results

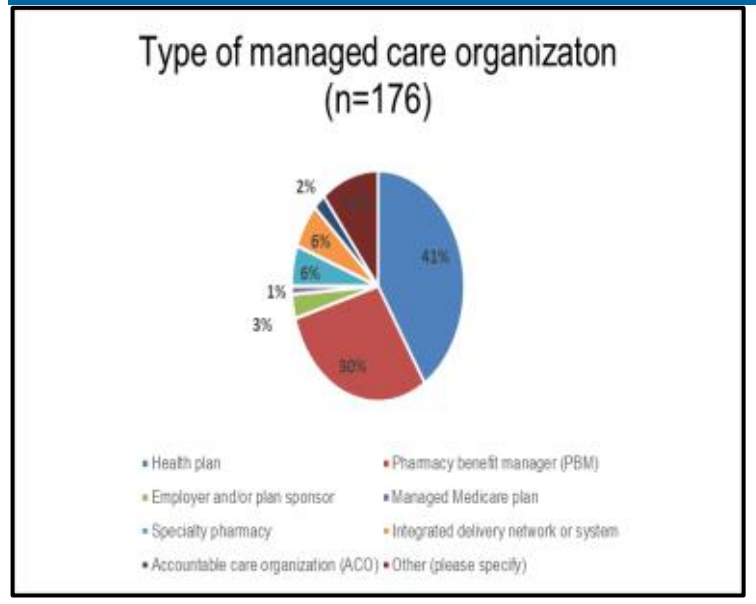

Type of managed care organizaton
(n=176)
2%
6%
6%
1%
3%
30%
41%
Health plan
Pharmacy benefit manager (PBM)
Employer and/or plan sponsor
Managed Medicare plan
Specialty pharmacy
Integrated delivery network or system
Accountable care organization (ACO)
Other (please specify)

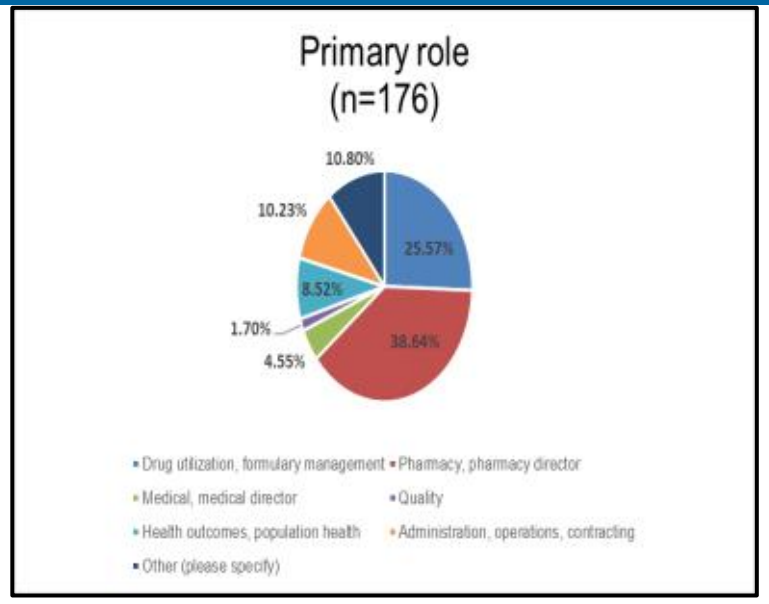

Primary role
(n=176)
10.80%
10.23%
25.57%
8.52%
1.70%
4.55%
38.64%
Drug utilization, formulary management
Pharmacy, pharmacy director
Medical, medical director
Quality
Health outcomes, population health
Administration, operations, contracting
Other (please specify)

48% of HCDMs reported that identifying meaningful surrogate endpoints linking to patient-centered outcomes early on in trials could minimize potential disconnects with payers in FDA expedited approvals.

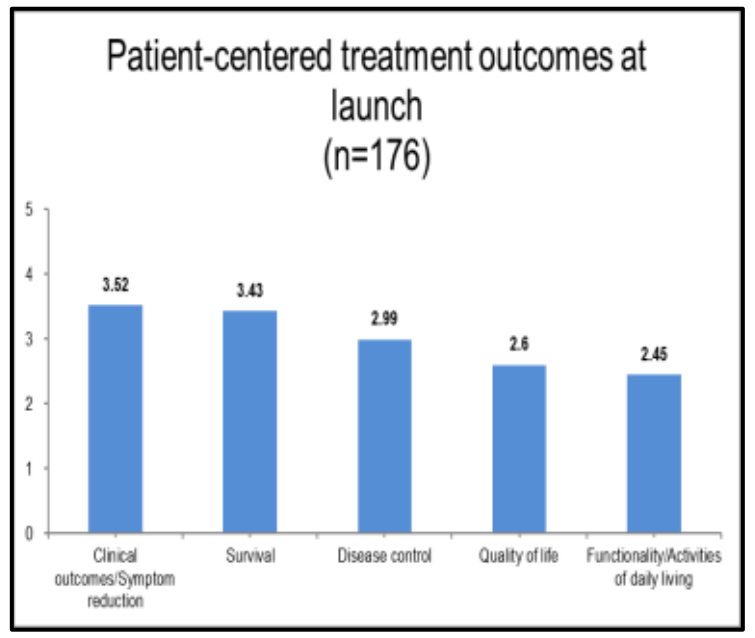

Patient-centered treatment outcomes at launch
(n=176)
5
4
3
2
1
0
3.52
3.43
2.99
2.6
2.45
Clinical outcomes/Symptom reduction
Survival
Disease control
Quality of life
Functionality/Activities of daily living

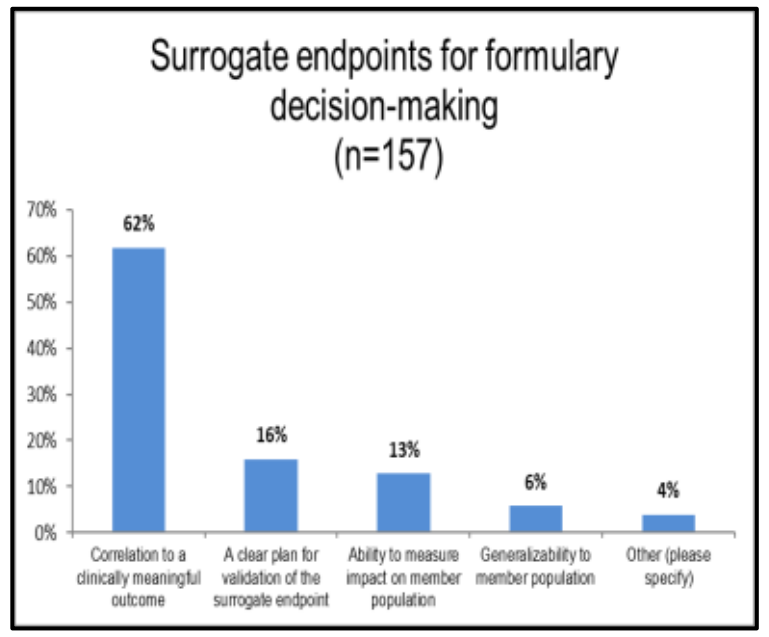

Surrogate endpoints for formulary decision-making
(n=157)
70%
60%
50%
40%
30%
20%
10%
0%
62%
16%
13%
6%
4%
Correlation to a clinically meaningful outcome
A clear plan for validation of the surrogate endpoint
Ability to measure impact on member population
Generalizability to member population
Other (please specify)

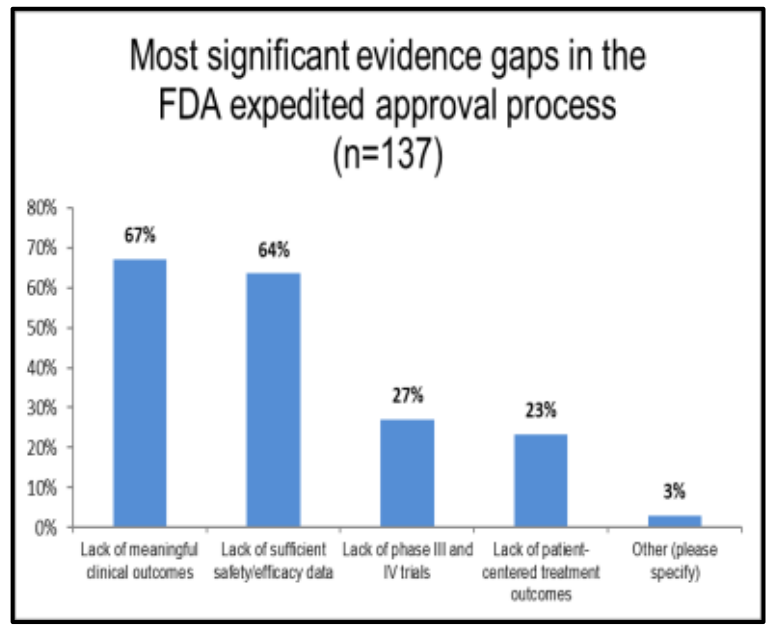

Most significant evidence gaps in the FDA expedited approval process
(n=137)
80%
70%
60%
50%
40%
30%
20%
10%
0%
67%
64%
27%
23%
3%
Lack of meaningful clinical outcomes
Lack of sufficient safety/efficacy data
Lack of phase III and IV trials
Lack of patient-centered treatment outcomes
Other (please specify)

Conclusions

- Majority of healthcare decision makers found that accelerated drug approvals are very impactful to the future of healthcare (n=90, 54%).
- Many did not have a separate and/or expedited review process, nor plans to create one.
- Lack of clinically meaningful outcomes was the top evidence gap in FDA expedited approvals.
- Health care decision makers reported that identifying meaningful surrogate endpoints that link to patient-centered treatment outcomes at an early point during trials could best minimize the potential disconnect with payers in FDA expedited approvals.
- Additionally, conducting phase III and phase IV trials more quickly could also minimize the potential disconnect with payers in the FDA expedited approvals process.
- Overall, the most important consideration in the formulary-decision making process is correlation to a clinically meaningful outcome.
- Future research will focus on performing an independent t-test to compare 2018-2021 cohort perspectives. Also, conventional content analysis will be conducted upon responses from open-ended questions.

Acknowledgements

Special thank you to my preceptors for being the best and most supportive mentors:
Dr. Annesha White, PharmD, MS, PhD, UNT
Dr. John Spain, MA, PharmD, BCPS, Pfizer, Inc.
Paula J Eichenbrenner, MBA, CAE, AMCP Foundation

References

1. Kesselheim, A., Wang, B., Franklin, J. and Darrow, J., 2021. Trends in Utilization of FDA Expedited Drug Development and Approval Programs, 1987-2014: cohort study.
2. CARROLL, J., 2021. The Accelerated Approval Debate: Faster FDA Drug Approvals May Mean Less Efficacy Data. [online] PubMed Central (PMC). Available at: <https://www.ncbi.nlm.nih.gov/pmc/articles/PMC3411233/>
3. Kaltenboeck, A., et al. 2021. Strengthening the Accelerated Approval Pathway. ICER. [online] Available at: <https://tinyurl.com/ICERAccel>
4. Evidera.com. 2021. [online] Available at <https://www.evidera.com/wp-content/uploads/2015/04/Avoiding-the-Fast-Track-Disconnect.pdf>
5. AMCP Foundation Trends in Healthcare. 2018. [online] Available at: <https://tinyurl.com/AMCPFTrendsReport>

Poster presented at AMCP Nexus, October 18 - October 21, 2021